Mechanism of NPRL2 gene therapy induced antitumor immunity in KRAS/STK11 mutant anti-PD1 resistant metastatic human non-small cell lung cancer (NSCLC)

AACR 2024
Poster# 1420

Ismail M Meraz[1], Mourad Majidi[1], Renduo Song[1], Meng Feng[1], Lihui Gao[1], Qi Wang[2], Jing Wang[2], Elizabeth Shpall[3], Jack A Roth[1]

Thoracic and Cardiovascular Surgery[1], [2]Department of Bioinformatics and Computational Biology, [3]Department of Stem Cell Transplantation, MD Anderson Cancer Center, Houston, TX

Abstract

NPRL2/TUSC4 is a tumor suppressor gene whose expression is reduced in many cancers including NSCLC. Restoration of NPRL2 induces cell cycle arrest and apoptosis. We investigated the antitumor immune responses to NPRL2 gene therapy in aPD1 aPD1-resistant KRAS/STK11^{mt} NSCLC in a humanized mouse model. Humanized mice were generated by transplanting human cord blood-derived CD34 stem cells into NSG mice. Mice harboring > 25% human CD45 cells were considered humanized. Lung metastases were developed in humanized mice by injecting KRAS/STK11^{mt}/aPD1^R A549 cells, which were treated with i.v. injection of NPRL2 nanoparticles (DOTAP-NPRL2) with or without pembrolizumab (aPD1). NPRL2 treatment reduced lung metastases, whereas pembrolizumab was ineffective. The antitumor effect was greater in humanized than non-humanized mice suggesting the role of immune response. The antitumor effect was associated with increased infiltration of human cytotoxic immune cells and decreased numbers of Treg in tumors. NPRL2+ pembrolizumab was not synergistic in this resistant model but was synergistic in KRAS^{wt}/aPD1^S H1299 tumors. Cytotoxic immune cells in tumors were associated with the antitumor effect. Consistent with A549, NPRL2 showed a significantly strong antitumor effect on another KRAS^{mt}/aPD1^R syngeneic LLC2 tumors whereas aPD1 was not effective. The antitumor effect of NPRL2 was correlated with HLA-DR$^+$ DC, CD11c DC, TILs, and NK cells in TME. The antitumor effect of NPRL2 was abolished upon in-vivo depletion of CD4, CD8 T, and MΦ in the LLC2 tumor model. However, no effect was found upon in-vivo depletion of NK cells. Nanostring analysis on lung metastasis resulted in a distinct pattern of human gene expression by NPRL2. T cell functional genes, including IFNγ, CD8b, CD7, TNFSF18, ITGA1, GATA3, and TBX21 were significantly increased. Conversely, the negative regulatory genes, including FOXP3, TGFB1, TGFB2, and IL-10RA were inhibited. NPRL2 also downregulated T cell co-inhibitory molecules, including CTLA4, ICOS, LAG3, PDCD1, CD274, IDO1, PDCD1LG2, CD47, and KLRB1. Stably expressing NPRL2 clones were established, and tumors in humanized mice with these clones exhibited significantly slower growth compared to controls. TME analysis showed an upregulation of human CD45, CD3, CD8 T, HLA-DR+ DC and a downregulation of Tregs, CD3$^+$PD1$^+$T, MDSC, and CD163$^+$ TAM in tumors expressing NPRL2. The stable cells showed a substantial increase in both colony formation inhibition and apoptosis. Stable clones showed heightened sensitivity to carboplatin in colony formation, apoptosis, and PARP cleavage assays. Stable expression of NPRL2 resulted in the downregulation of both AKT-mTOR and MAPK pathways by inhibition of pAKT, pmTOR, pPRAS40, p4E-BP1, pS6, and pERK1/2. Taken together, NPRL2 gene therapy induces antitumor activity on KRAS/STK11^{mt}/aPD1^R tumors through DC-mediated antigen presentation and cytotoxic immune cell activation.

Methods: NPRL2 Gene Therapy in Humanized Mice

- KRAS/STK11 mutant A549 and KRAS WT H1299 cells were transfected with NPRL2 and apoptosis & inhibition of colony formation were measured.
- Humanized mice were generated by transplanting fresh human cord blood derived CD34 stem cells into sub- lethally irradiated NSG mice. The level of engraftment of human CD45, CD3 T, CD19 B, NK cells was verified before tumor implantation. Mice harboring > 25% human CD45 cells were considered humanized.
- KRAS/STK11 mutant anti-PD1 resistant A549 NSCLC cells were injected intravenously into fully humanized NSG mice and developed lung metastasis. Metastases were treated with intravenous injection of NPRL2 gene loaded cationic lipid nanoparticles (DOTAP-NPRL2) with or without pembrolizumab (anti-PD1).

NPRL2 Inhibited Colony Formation And Induced Apoptosis

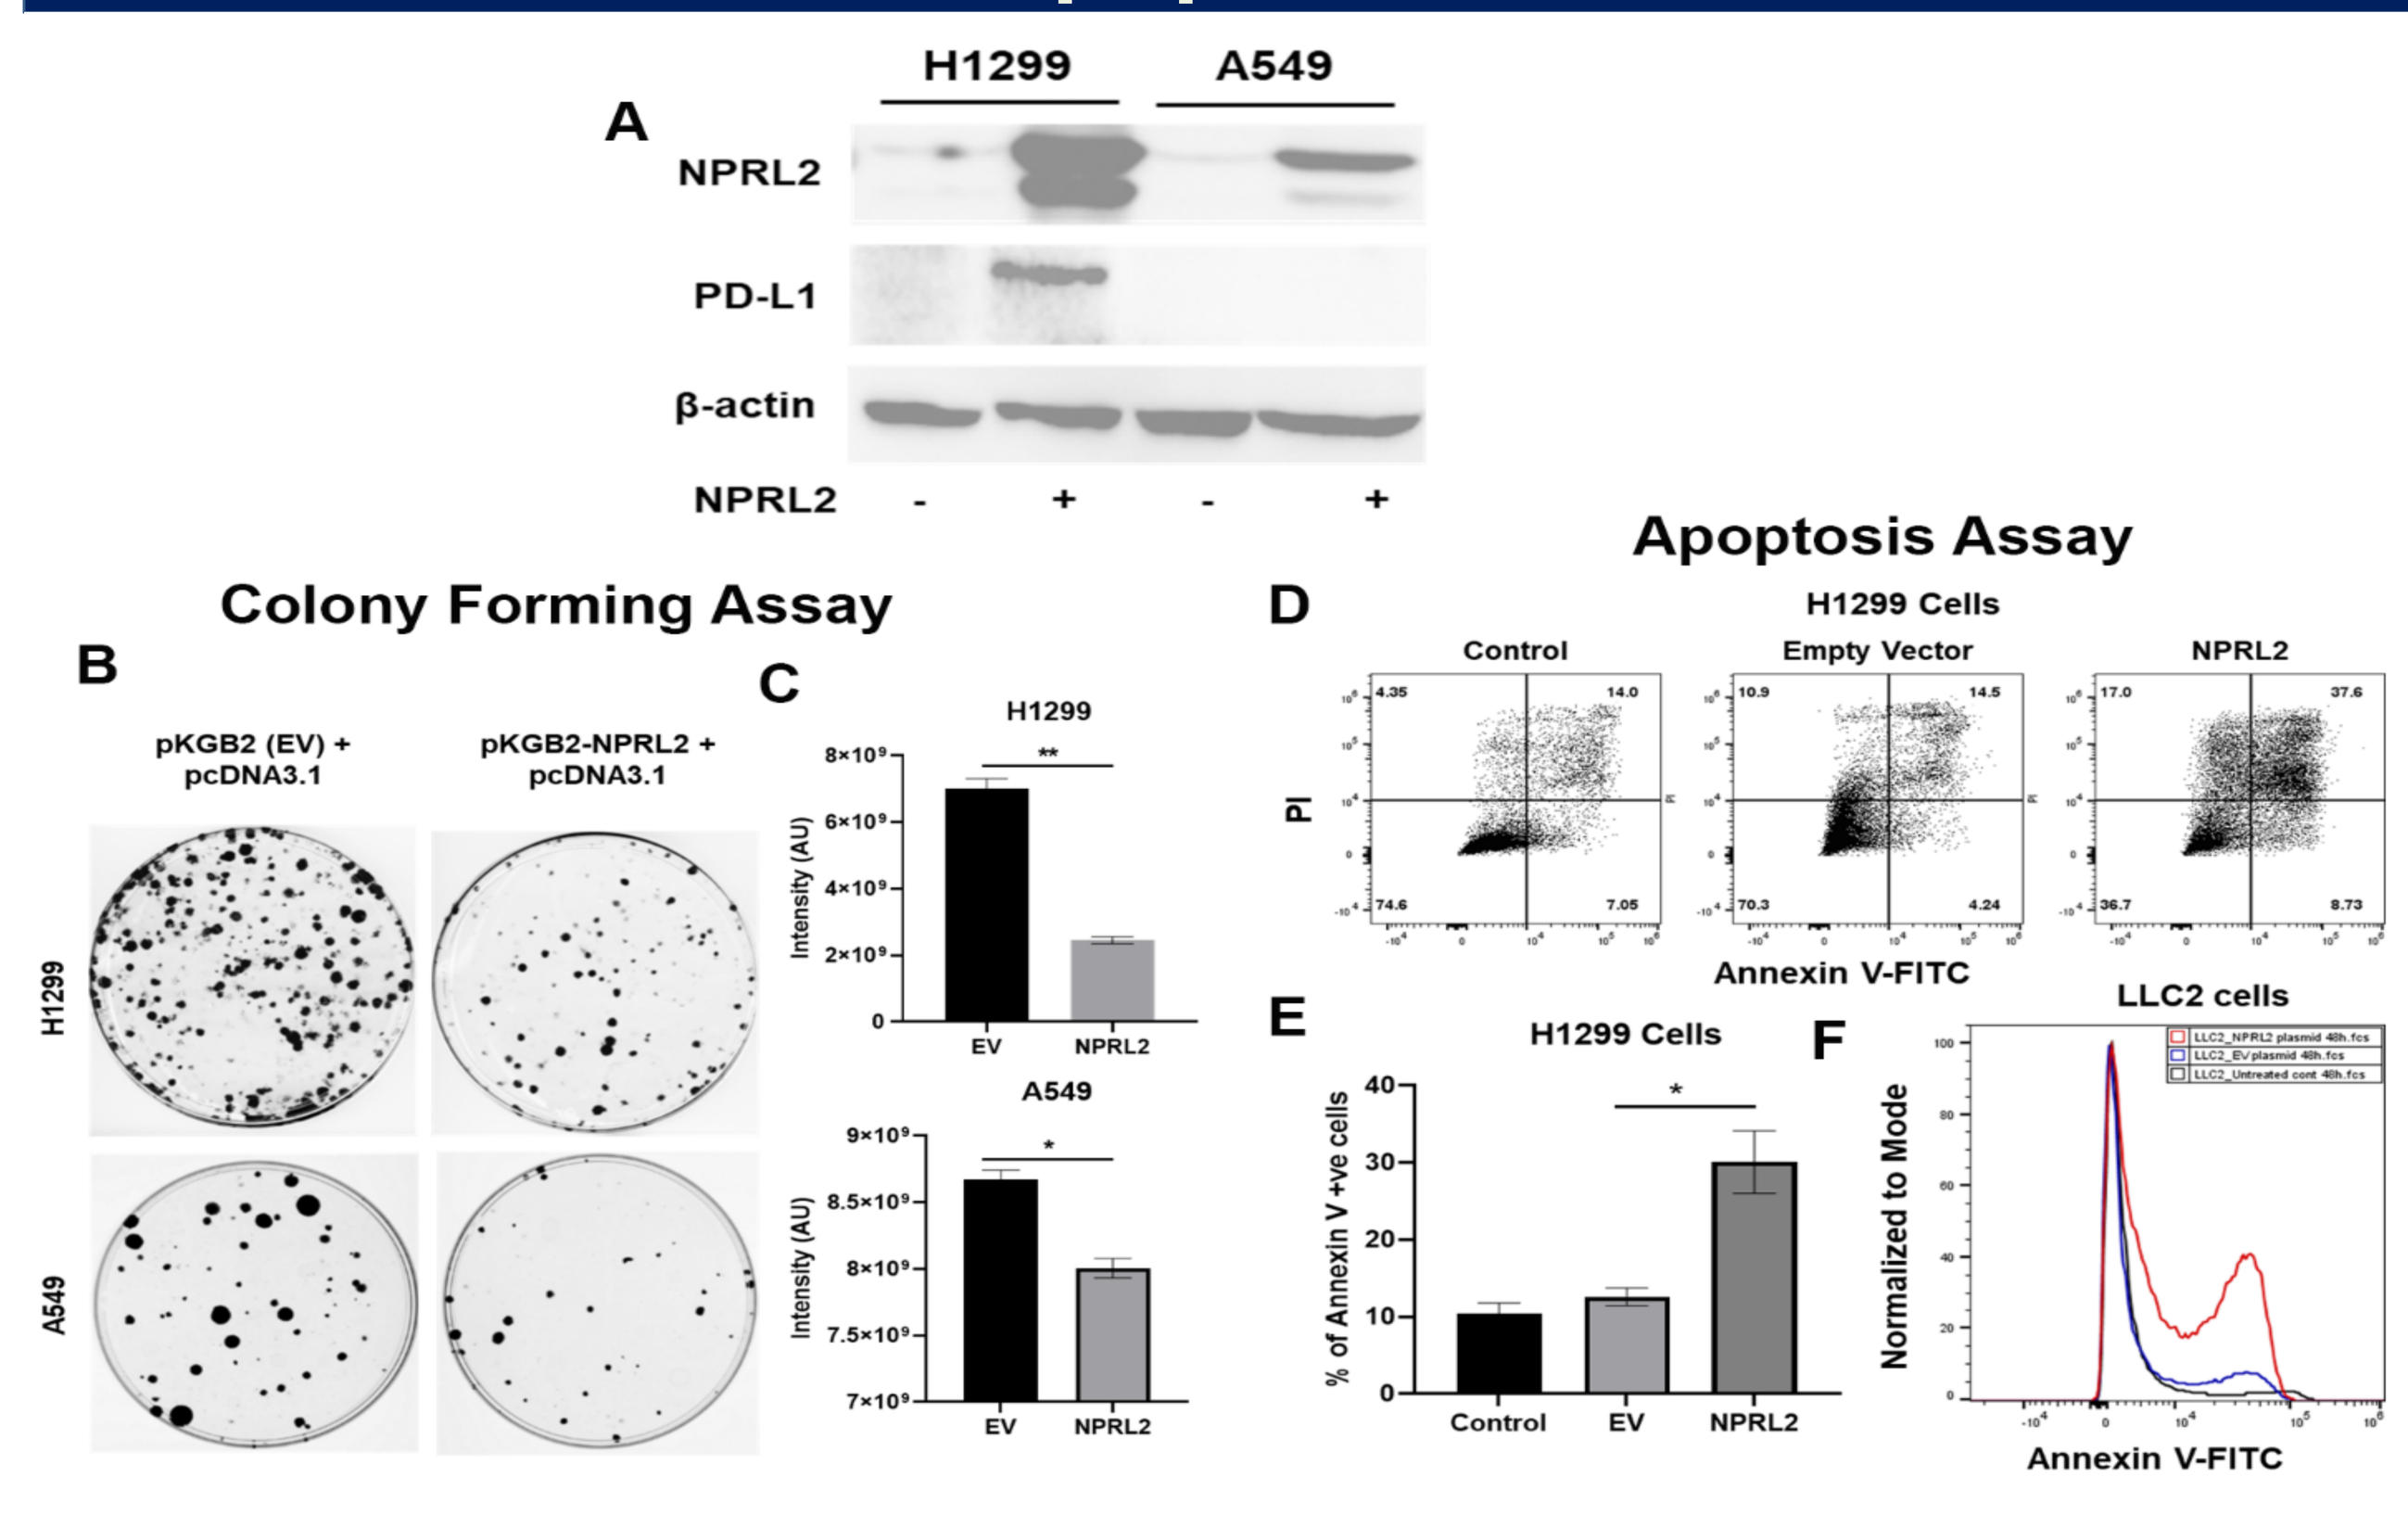

Fig 1. NPRL2 inhibited colony formation and induced apoptosis in NSCLC. A) Expression of NPRL2 and PD-L1 on H1299 and A549 cells after transfection; B-C) Colony formation assay and its quantitation on stably NPRL2 transfected H1299 and A549; D-F) Apoptosis on H1299 and LLC2 cells after NPRL2 transfection. * means p < 0.05

Antitumor Effect Of NPRL2 On KRAS^{mt}/STK11^{mt}/aPD1^R A549 Lung Metastases

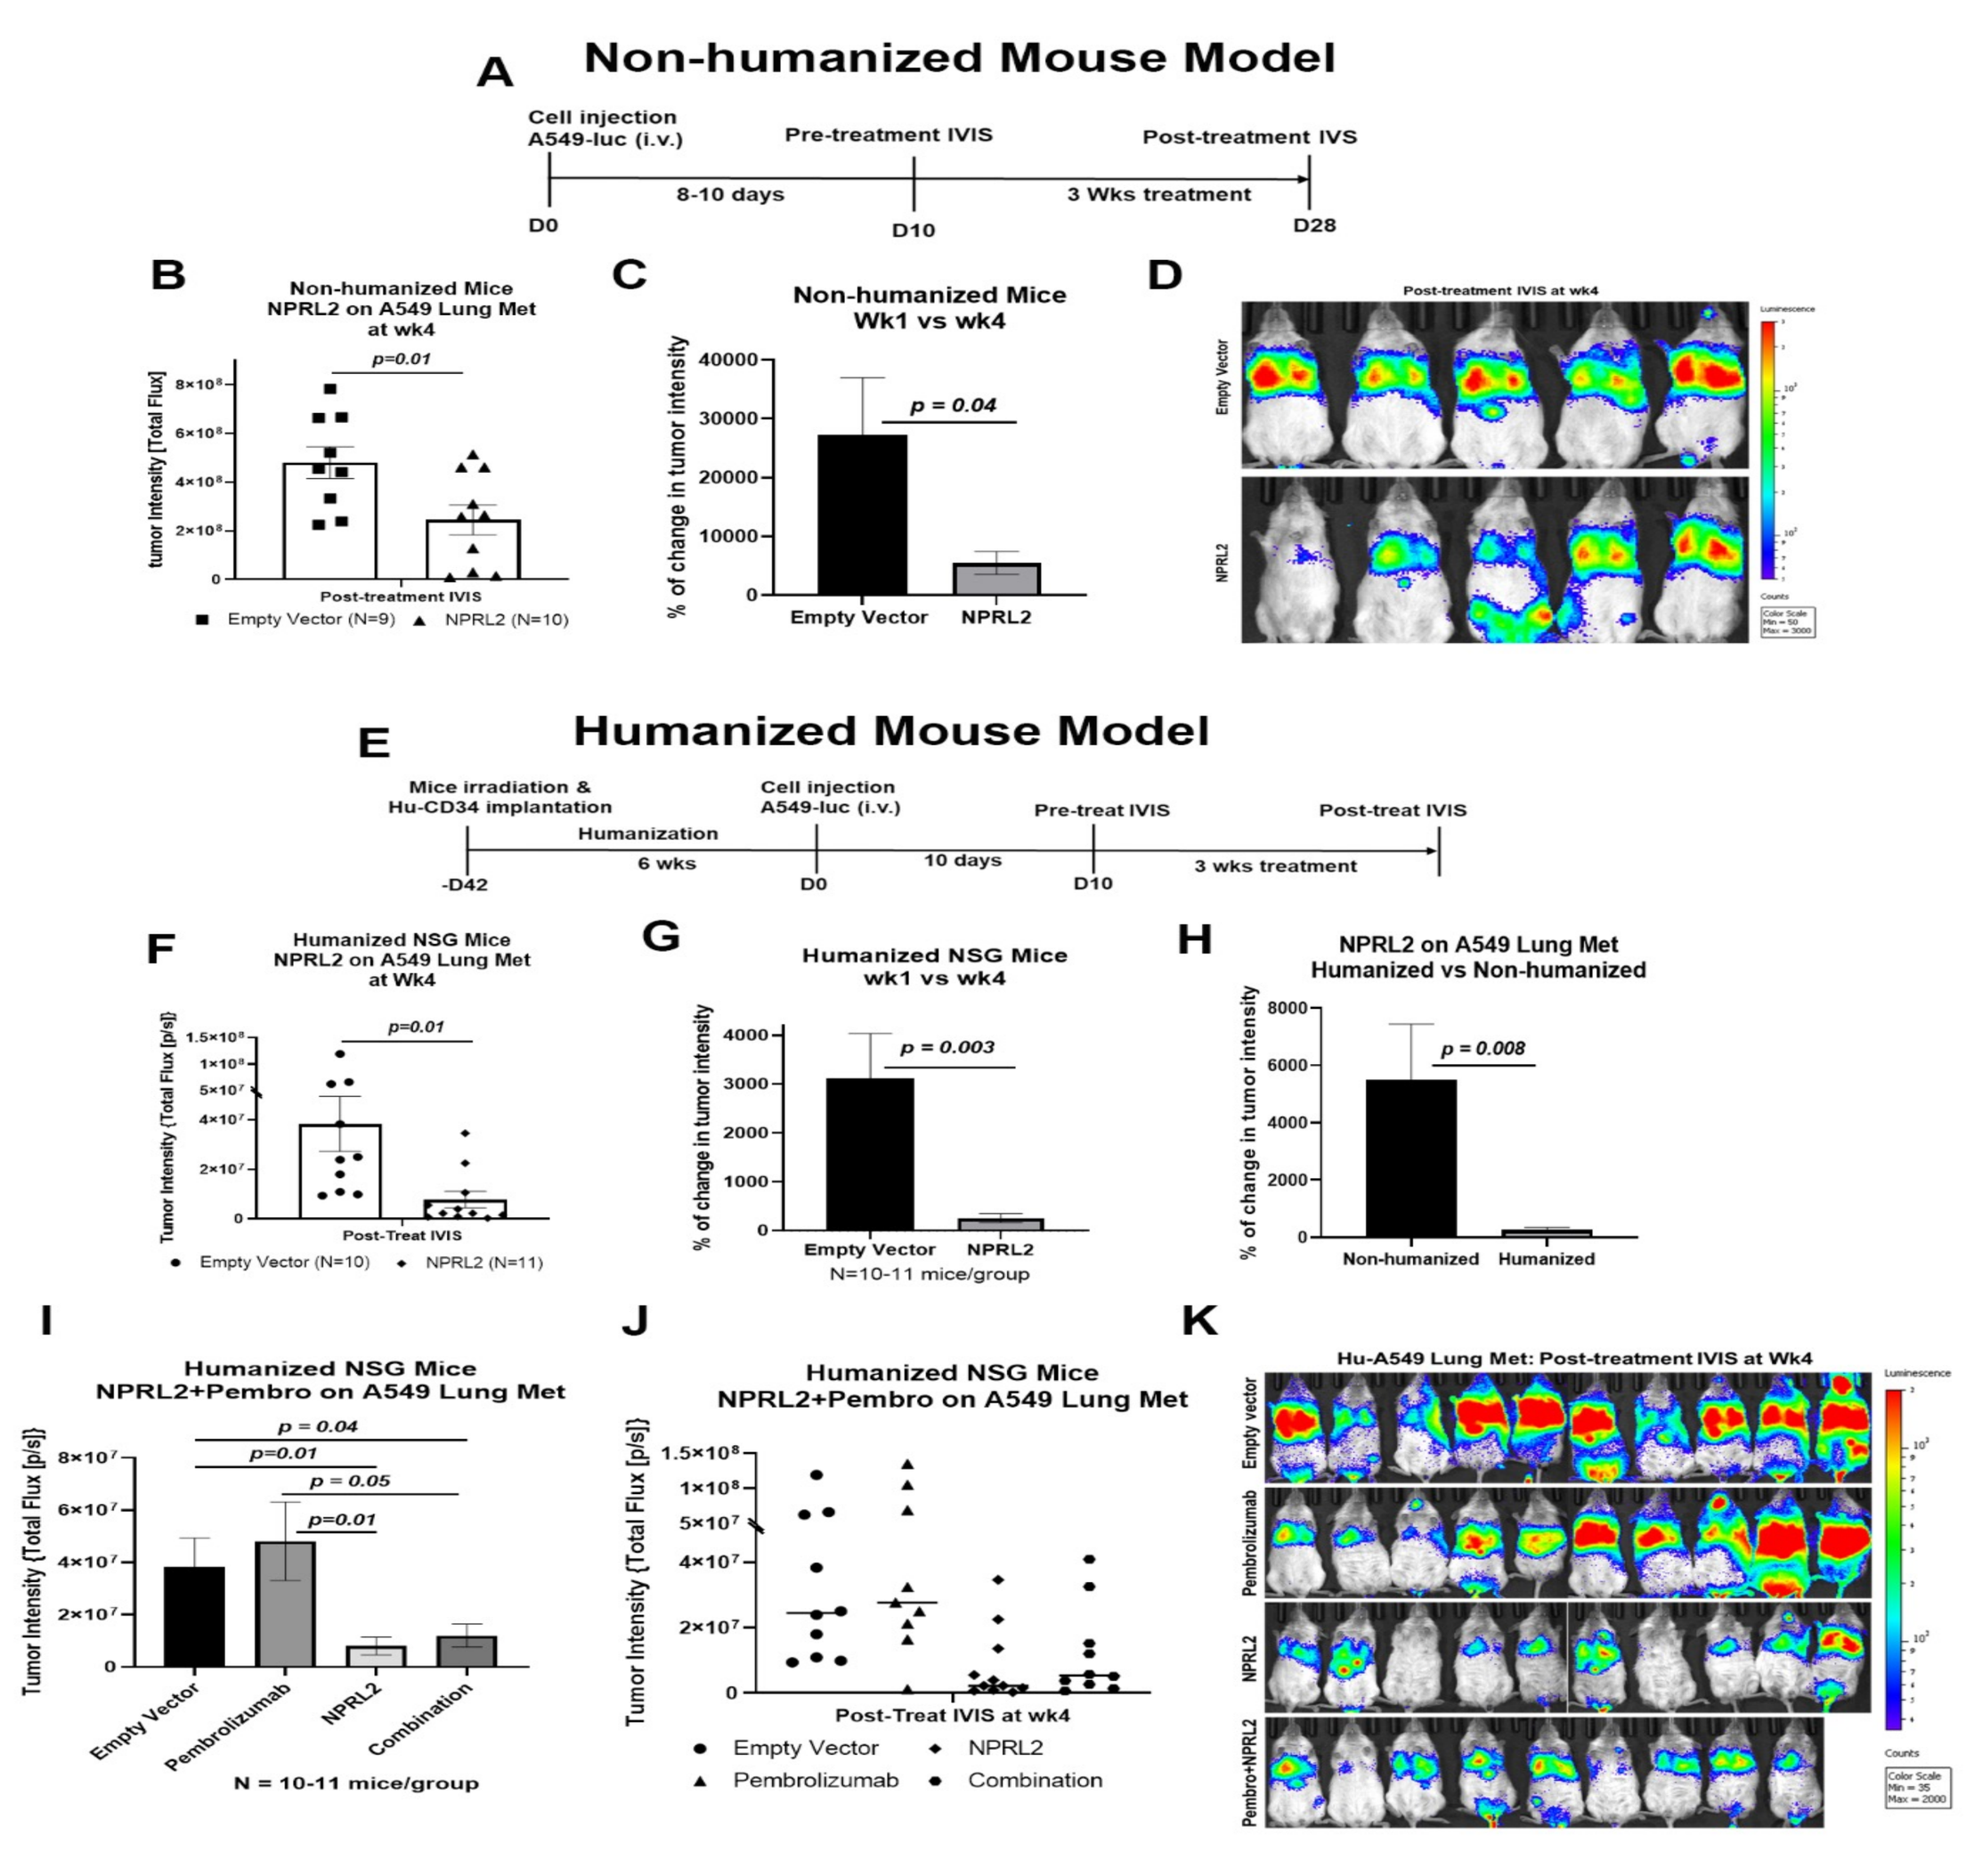

Fig 2. Antitumor effect of NPRL2 on anti-PD1 resistant KRAS/STK11 mutant A549 lung metastasis. A-D) NPRL2 effect on Non-Humanized Mice: A) treatment strategy, B) Tumor intensity, C) Rate of tumor burden reduction, D) Bioluminescence imaging on mice; E-K) NPRL2 effect on humanized mice: E) treatment strategy, F) NPRL2 effect on total tumor intensity, G) Percent of change in tumor intensity, H) comparison of tumor burden in Humanized vs Non-humanized system, I) effect of NPRL2+ Pembrolizumab on A549 lung metastasis, J) individual mouse response to treatment, K) IVIS imaging

NPRL2 Induces Antitumor Immune Responses In A Humanized Mouse Model

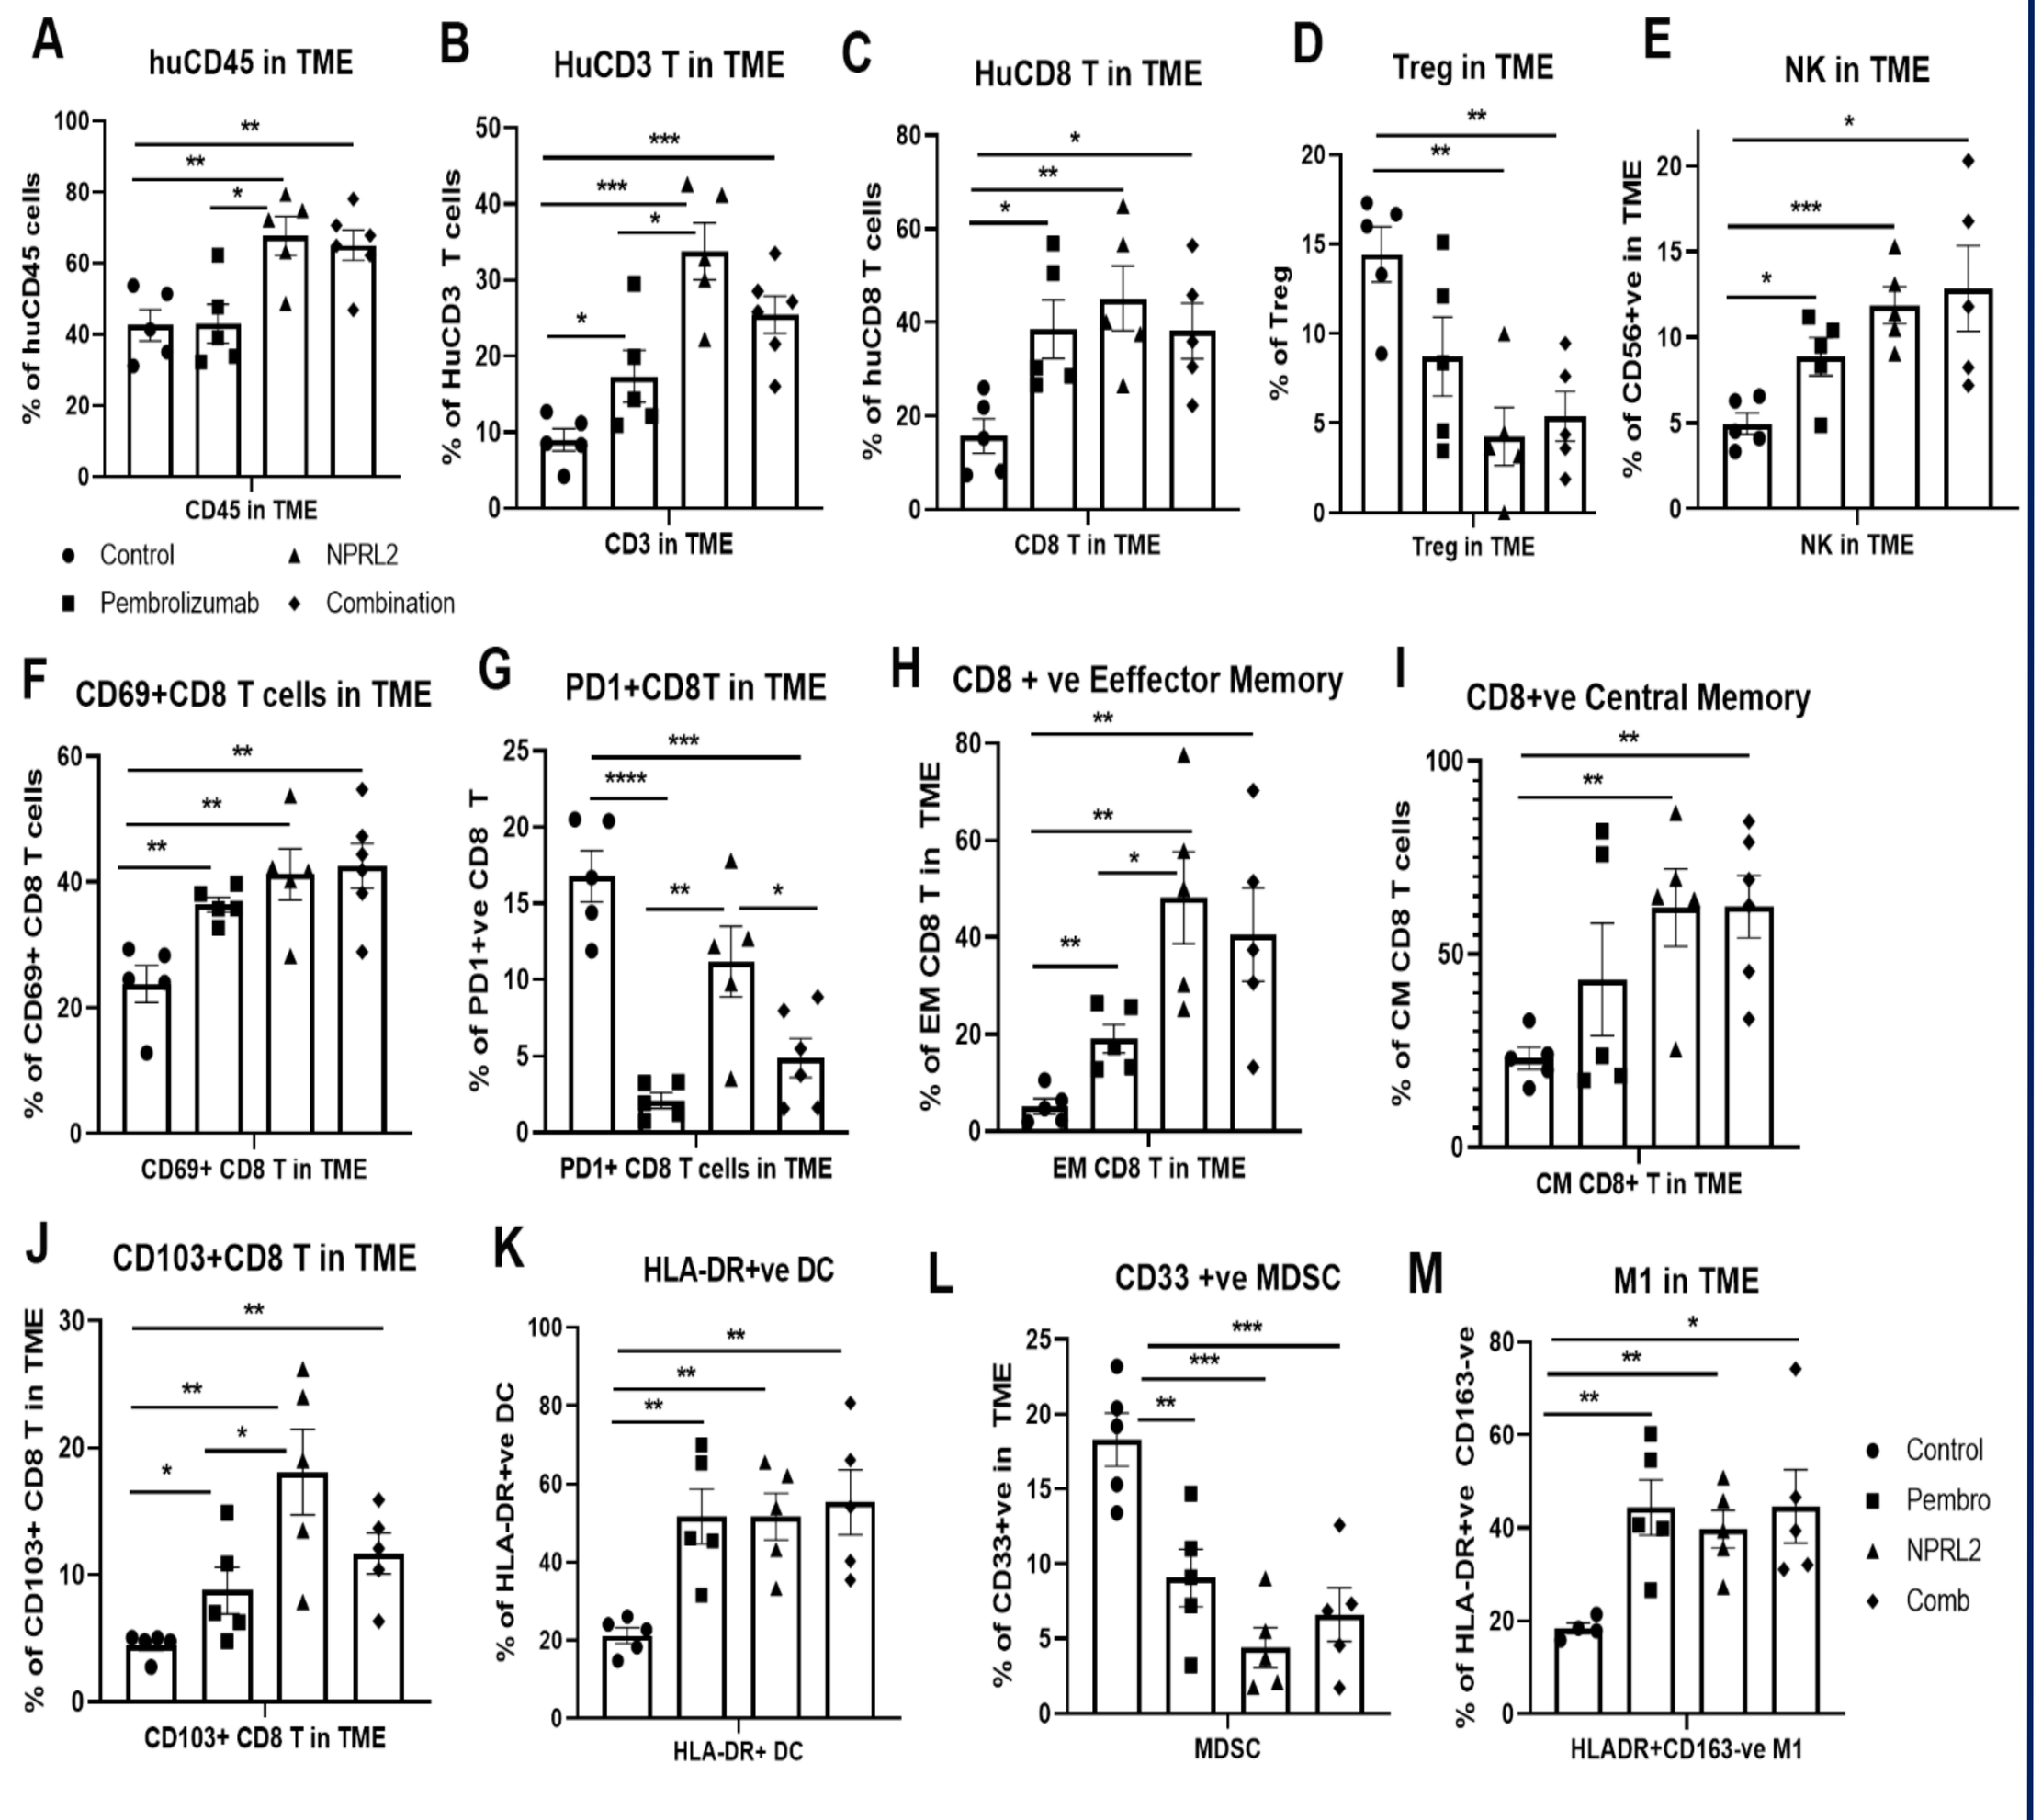

Fig 3. NPRL2 induces antitumor immune responses in A549-Lung Met in humanized mice. Lung met bearing humanized mice were treated with NPRL2 and its combination with pembrolizumab followed by TME analysis by flow cytometry. A-M) shows the levels of different adaptive immune cells such as CD3 T, CTL, Treg, NK, activated CTL, PD1 expressing CTL, effector and central memory T cells and innate immune cells such as DC, MDSC, M1 macrophages in lung mets. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

Synergistic Antitumor Effect Of NPRL2 + Pembrolizumab On KRAS^{wt}/aPD1^S H1299 Tumors In A Humanized Mouse Model

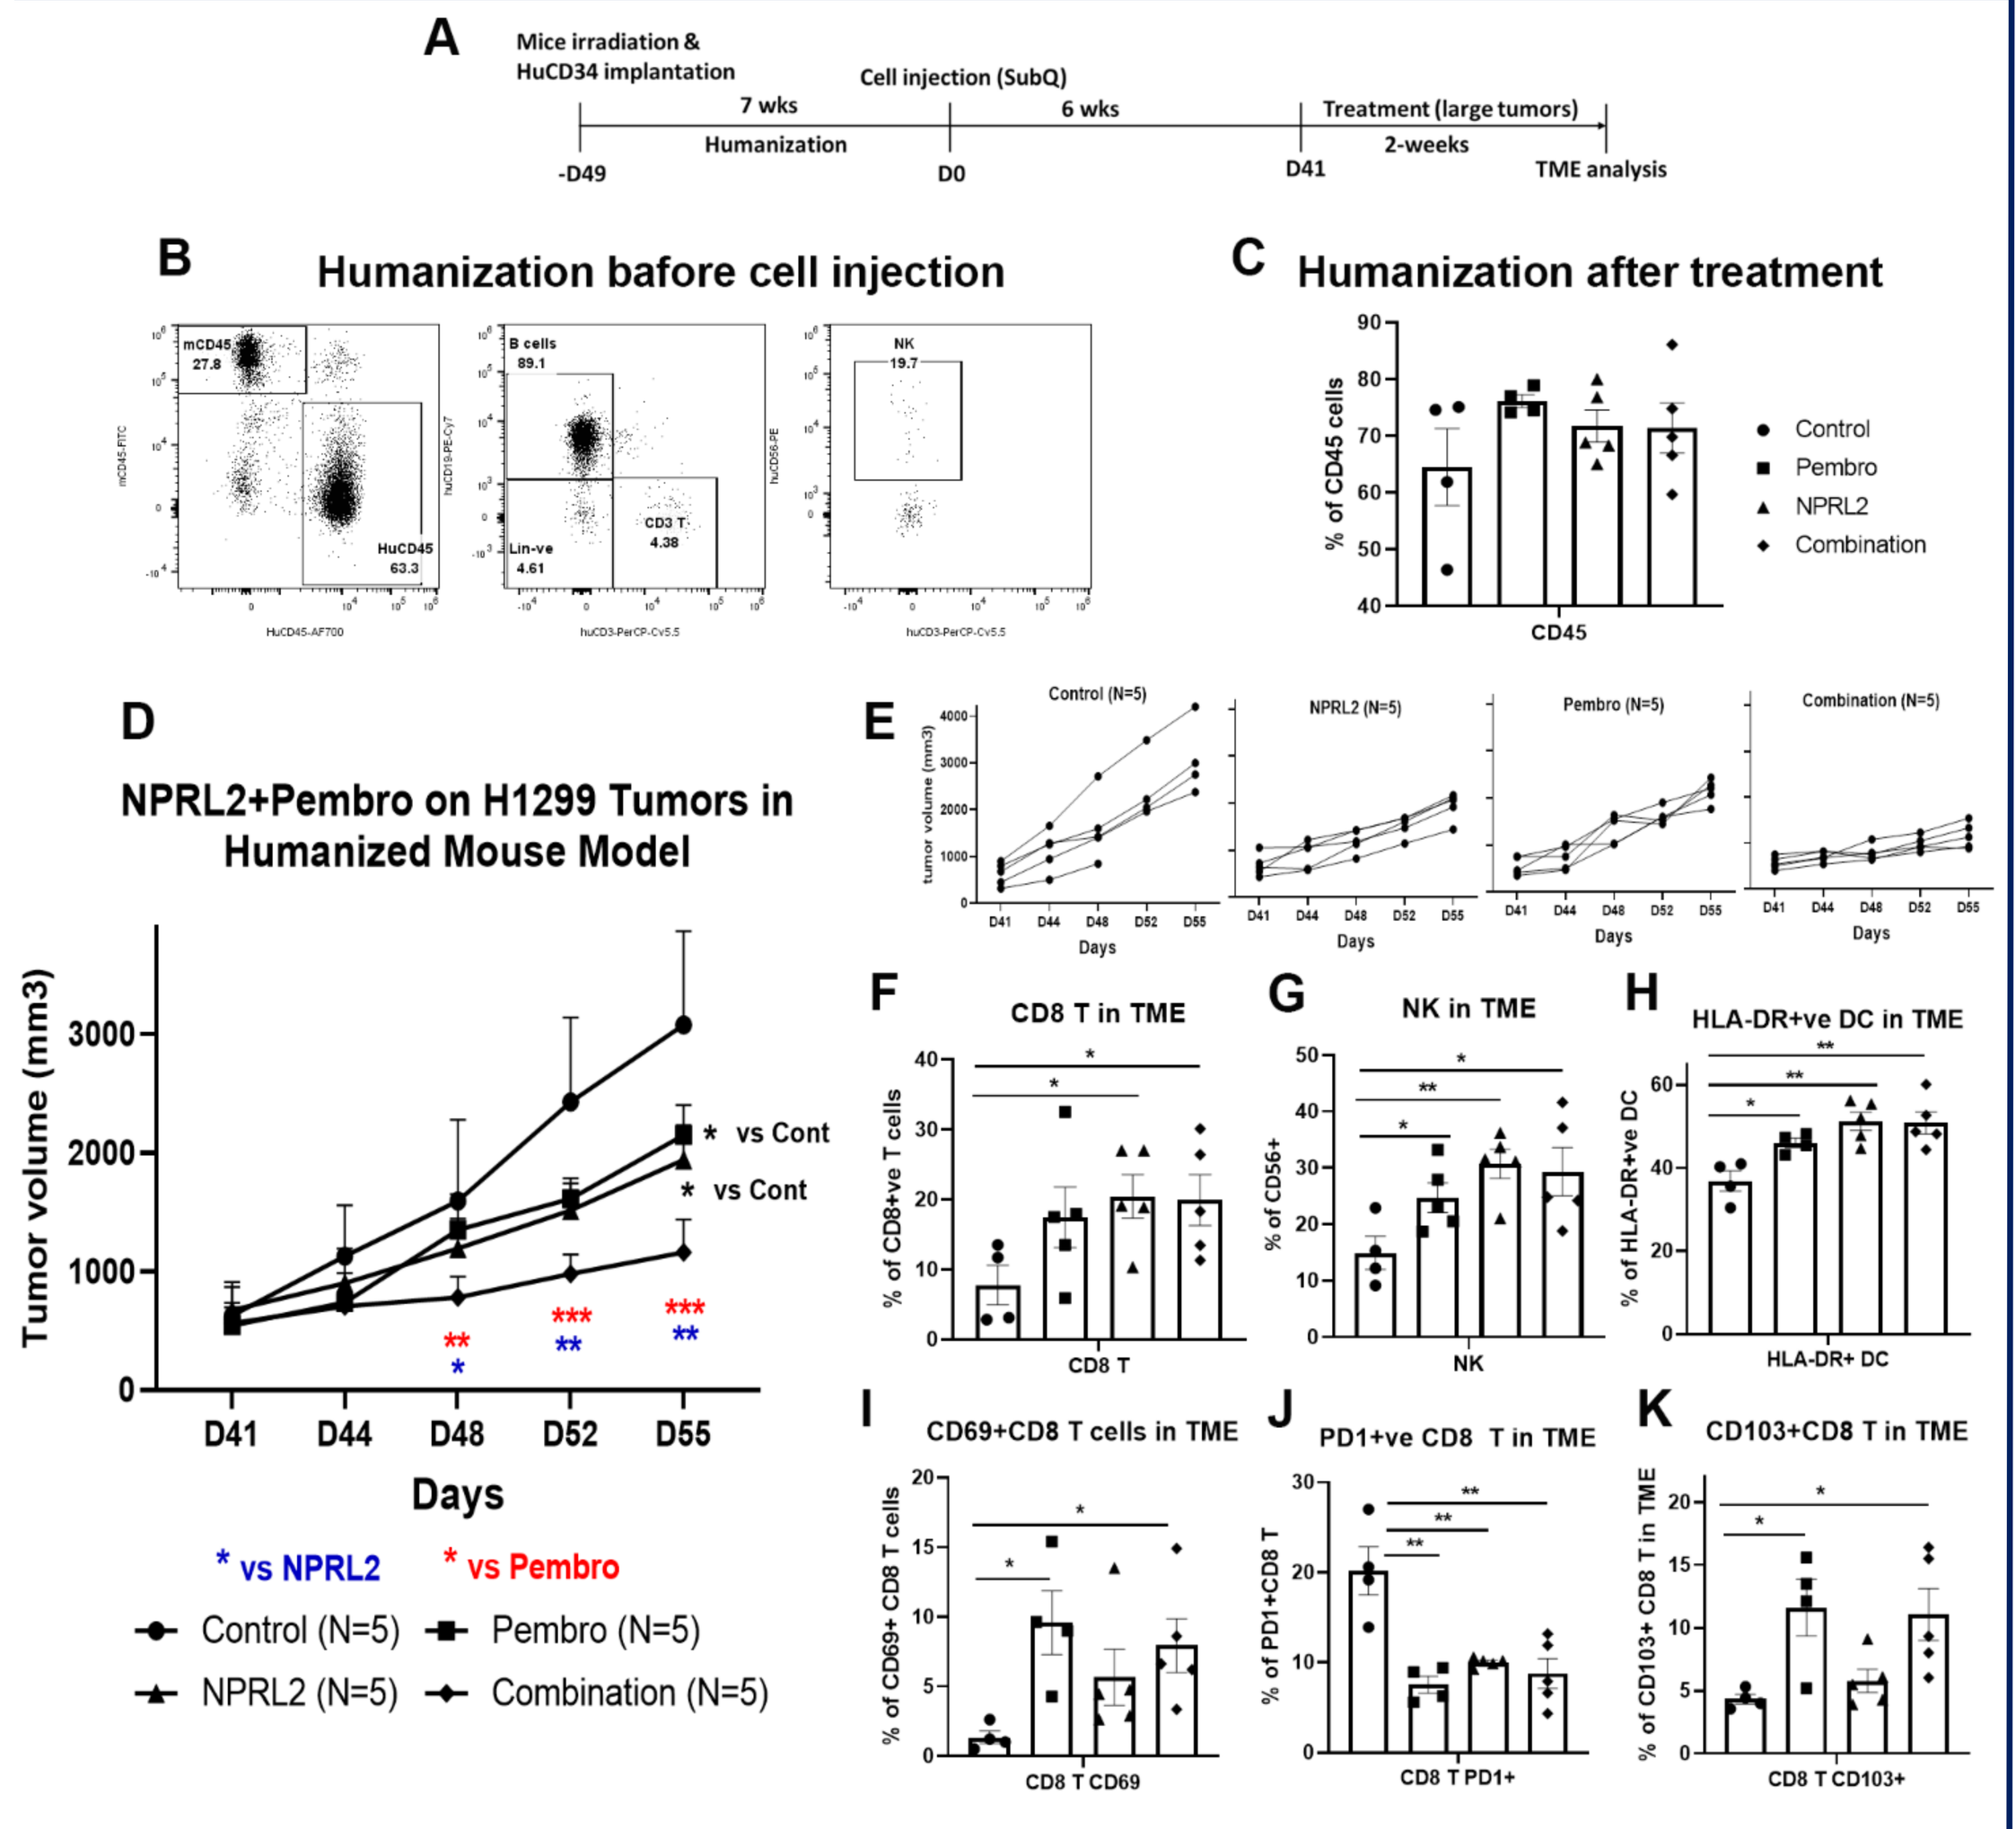

Fig 4. Synergistic antitumor effect of NPRL2 + Pembrolizumab on H1299 tumors in humanized mouse model. A) Humanization and treatment strategy, B) level of humanization status before experiment, C) Level of human CD45 cells after experiment, D) synergistic antitumor effect of the combination, E) individual mouse response to the treatments, F-K) TME analysis to show the level of infiltrating immune cells like CD8 T, NK cells, DC and activated CD8 T, PD1 expressing CD8 T in the tumors. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

NPRL2 Effect On KRAS^{mt}/aPD1^R LLC2 Tumors In A Syngeneic Mouse Model

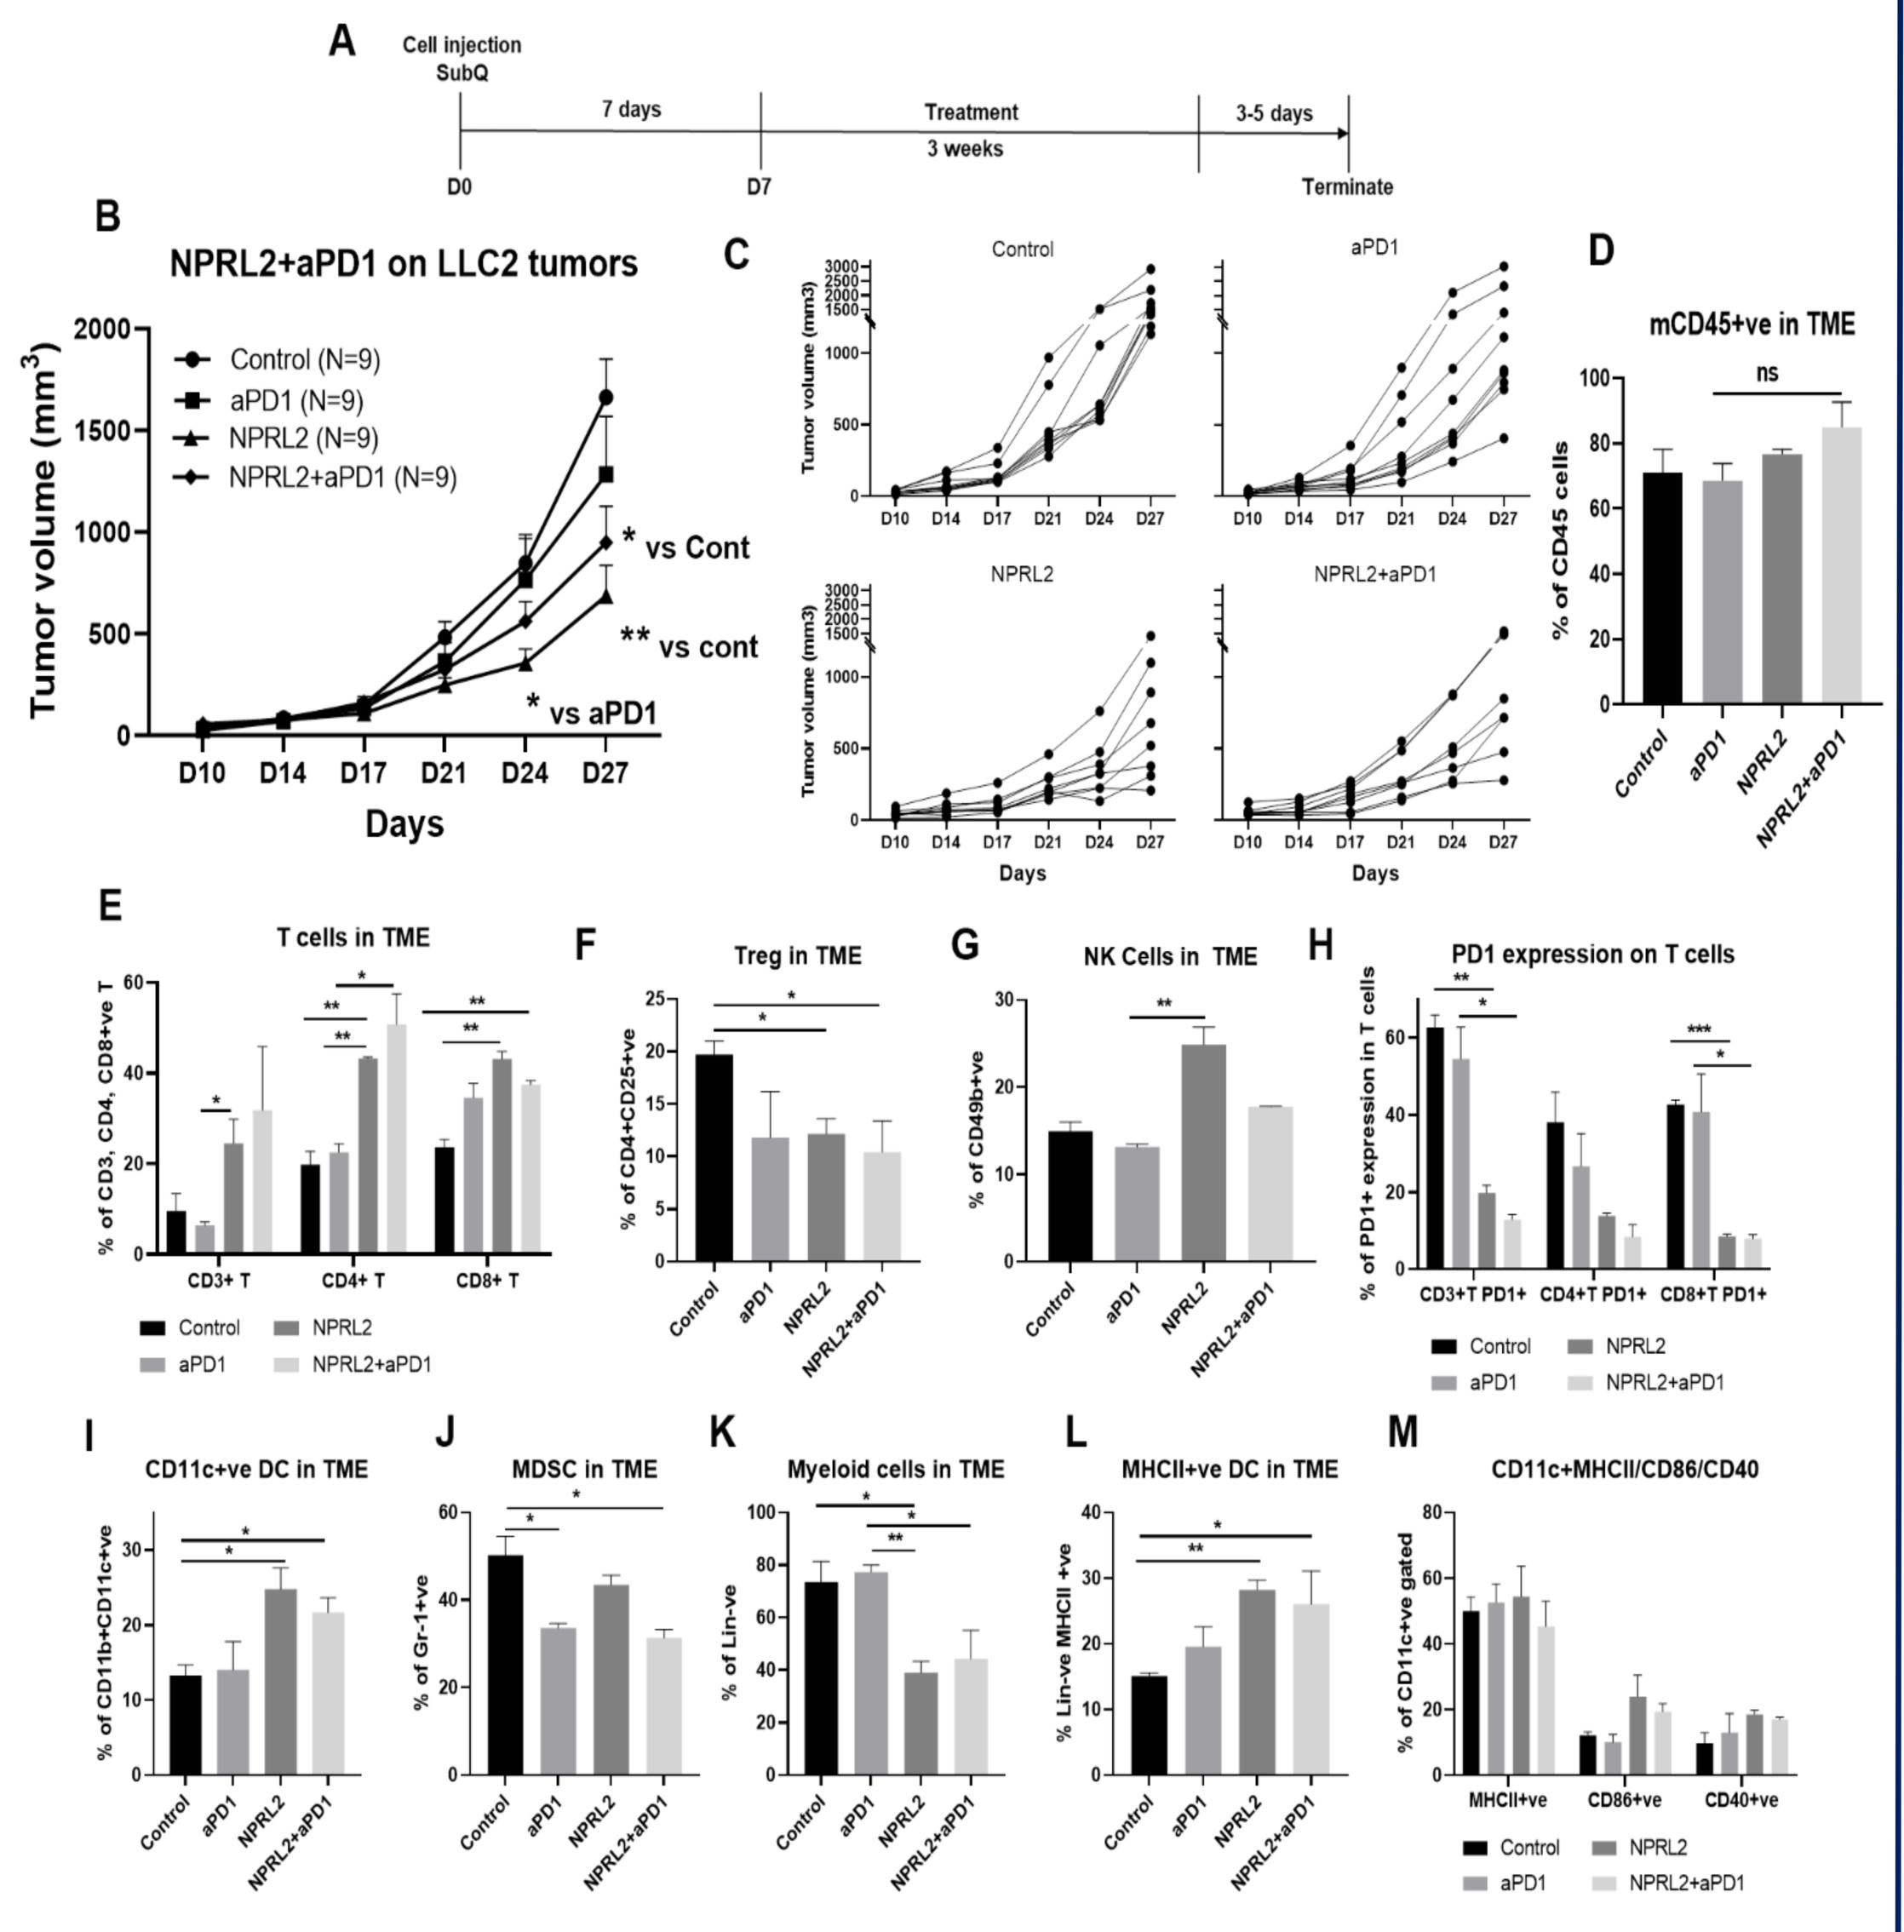

Fig 5. Antitumor effect of NPRL2 on anti-PD1 resistant KRAS mutant LLC2 tumors in syngeneic mice. A) treatment strategy, B) NPRL2 effects on LLC2 tumors, C) individual mouse response towards treatments, D-M) TME analysis to show the level of infiltrating immune cells in tumors. * means p < 0.05, ** means p<0.005, and *** means p < 0.0005

NPRL2 Antitumor Effect Is Dependent On Cytotoxic T Cells and Antigen Presenting Cells

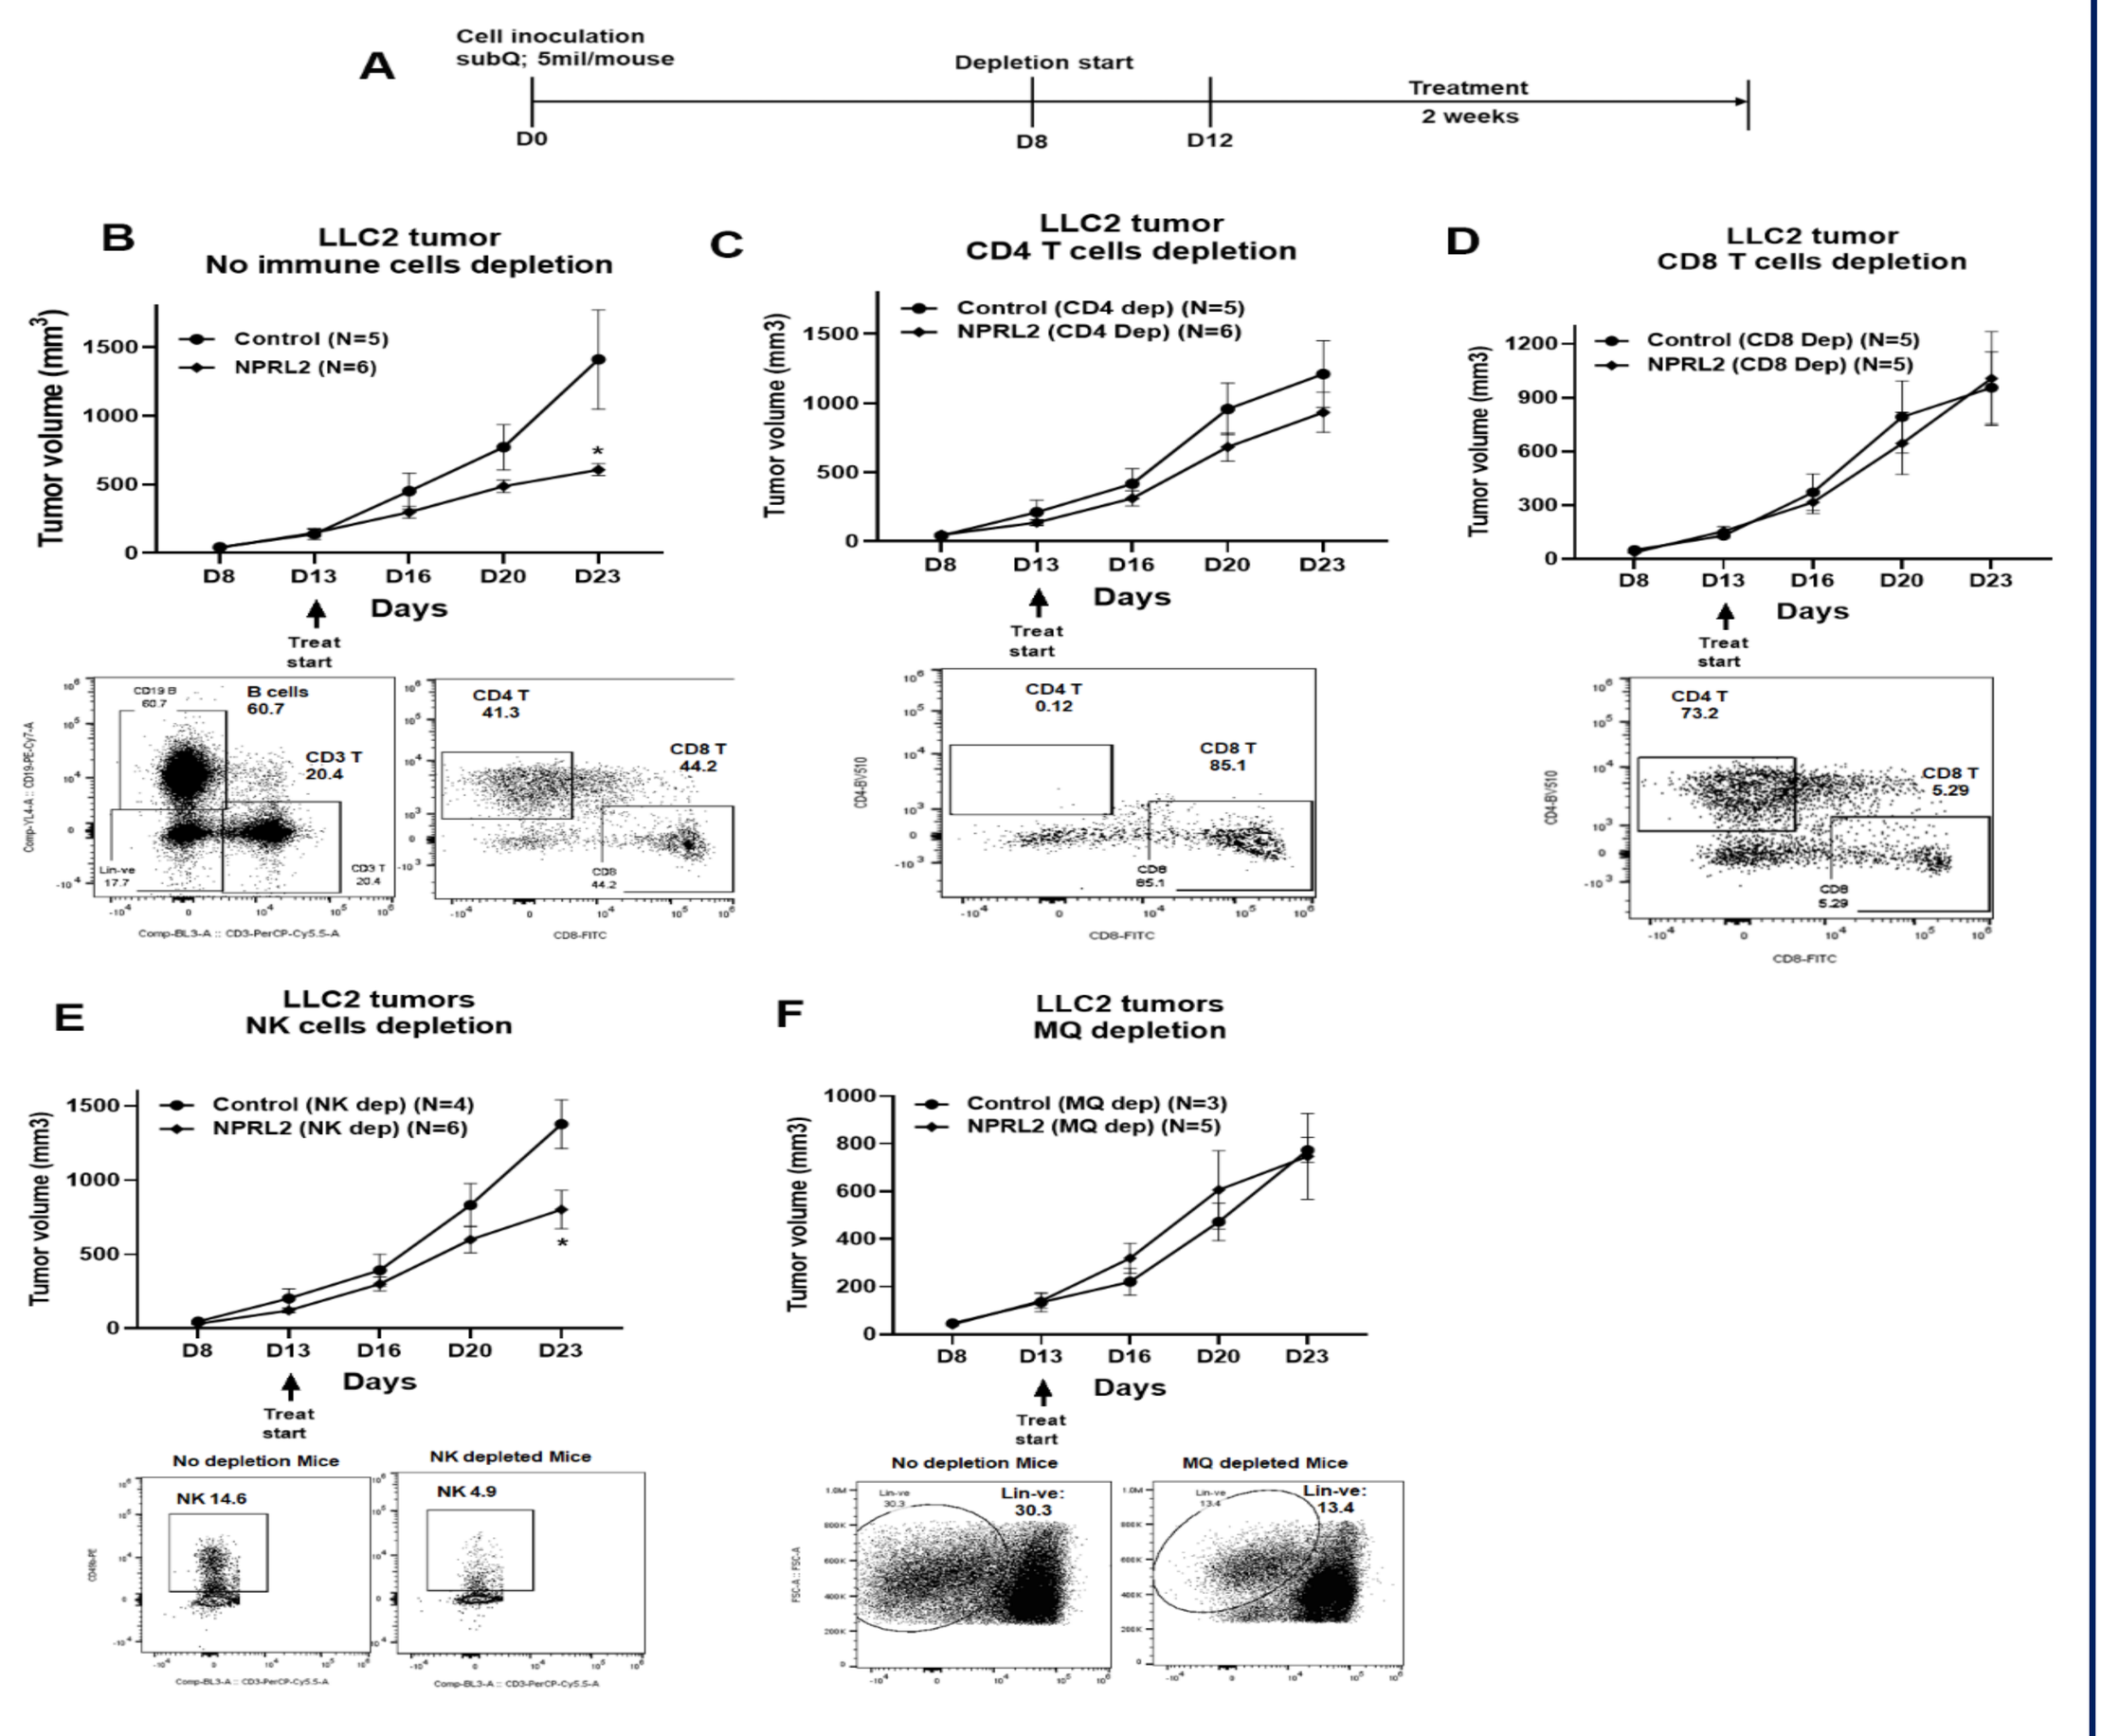

Fig 6. Antitumor effect of NPRL2 on specific immune cells depleted mice. CD4 T, CD8 T, NK and MQ cells were depleted from LLC2 tumor-bearing mice followed by NPRL2 treatment. A) Treatment strategy, B-F) Antitumor effect of NPRL2 on B) No immune cell-depleted, C) CD4 T cells depleted, D) CD8 T cells depleted, E) NK cells depleted, and F) MQ depleted mice. * means p < 0.05

NPRL2 Altered Gene Expression In Tumors Associated With T Cell Activation

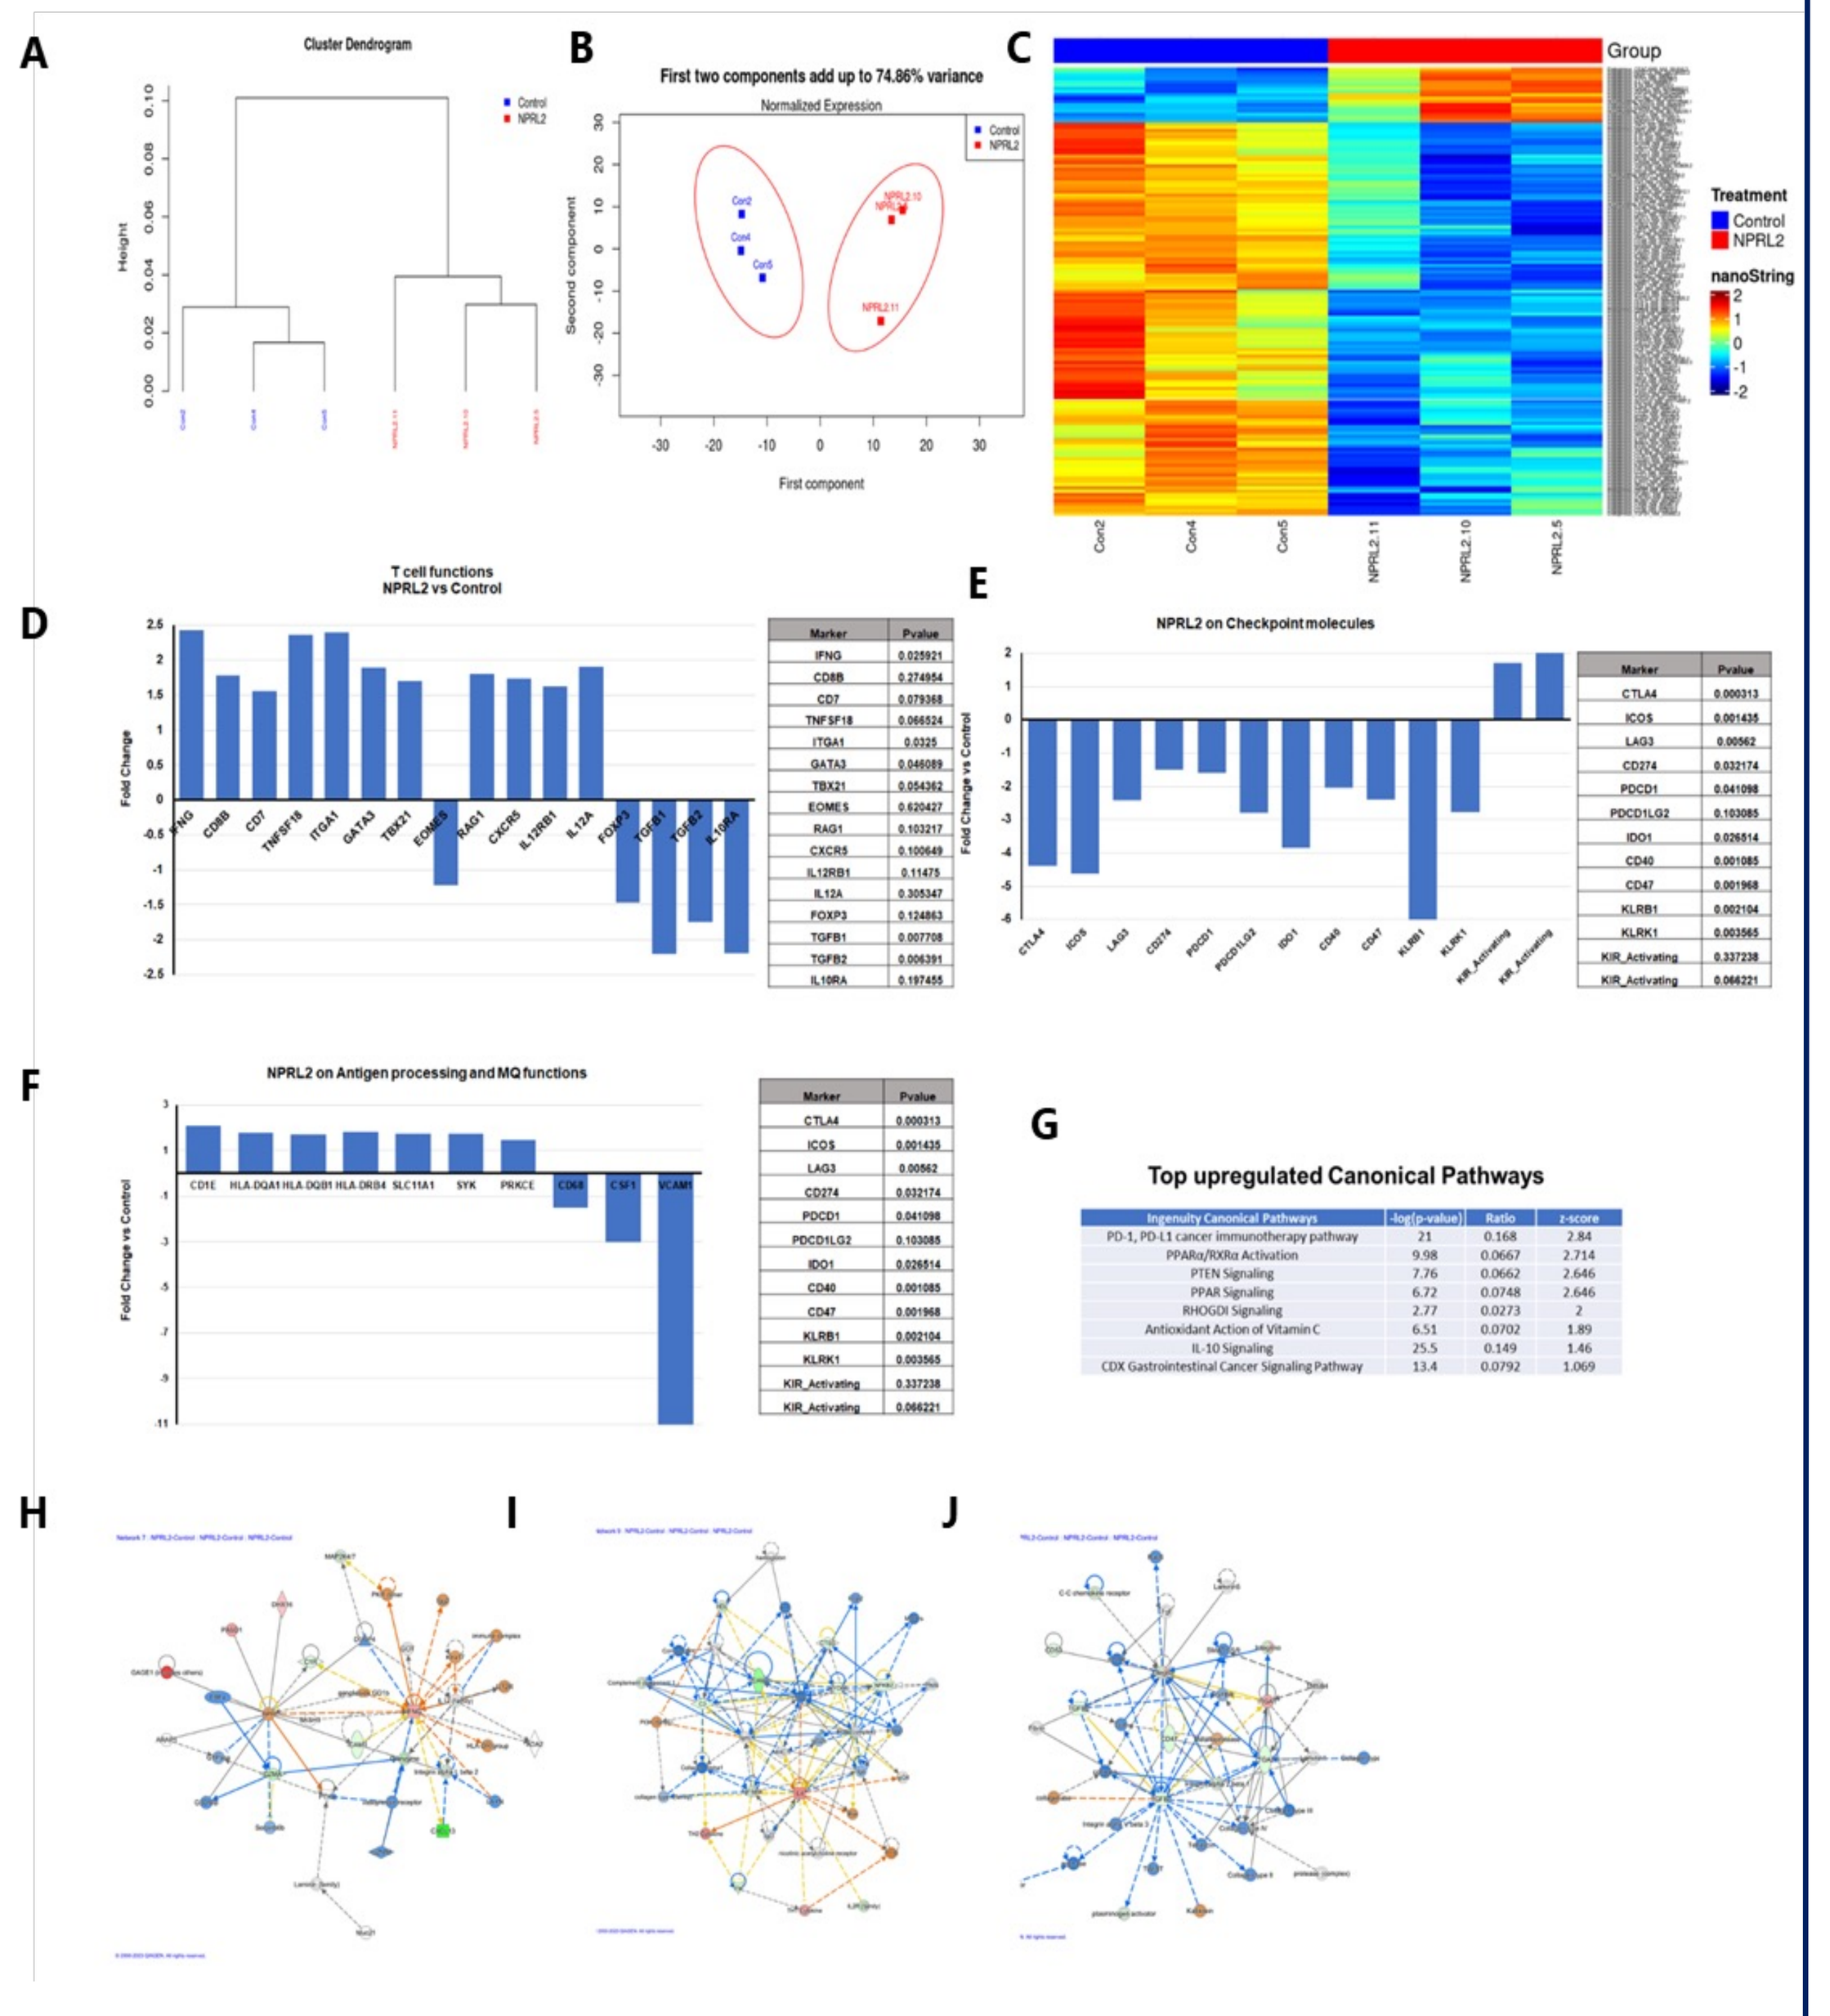

Fig 7. Alteration of gene expression associated with T cell activation by NPRL2 in humanized mice. Nanostring analysis for 770 human immune-related genes on NPRL2 treated tumors. A-B) Dendrogram and Principal Component curve shows the differences/distances among samples; C) Gene expression heat map; D) Expression of T cell activation genes; E) Checkpoint markers downregulated by NPRL2; F) Gene expression associated with Ag presentation; G) A list of top upregulated signaling pathways in Ingenuity pathway analysis; H-J) IFNγ, IL-4, and TGFβ signaling networks significantly altered by NPRL2

Permanent Restoration Of NPRL2 Altered TME, Induced Apoptosis, Inhibited Cell Growth And Its Signaling

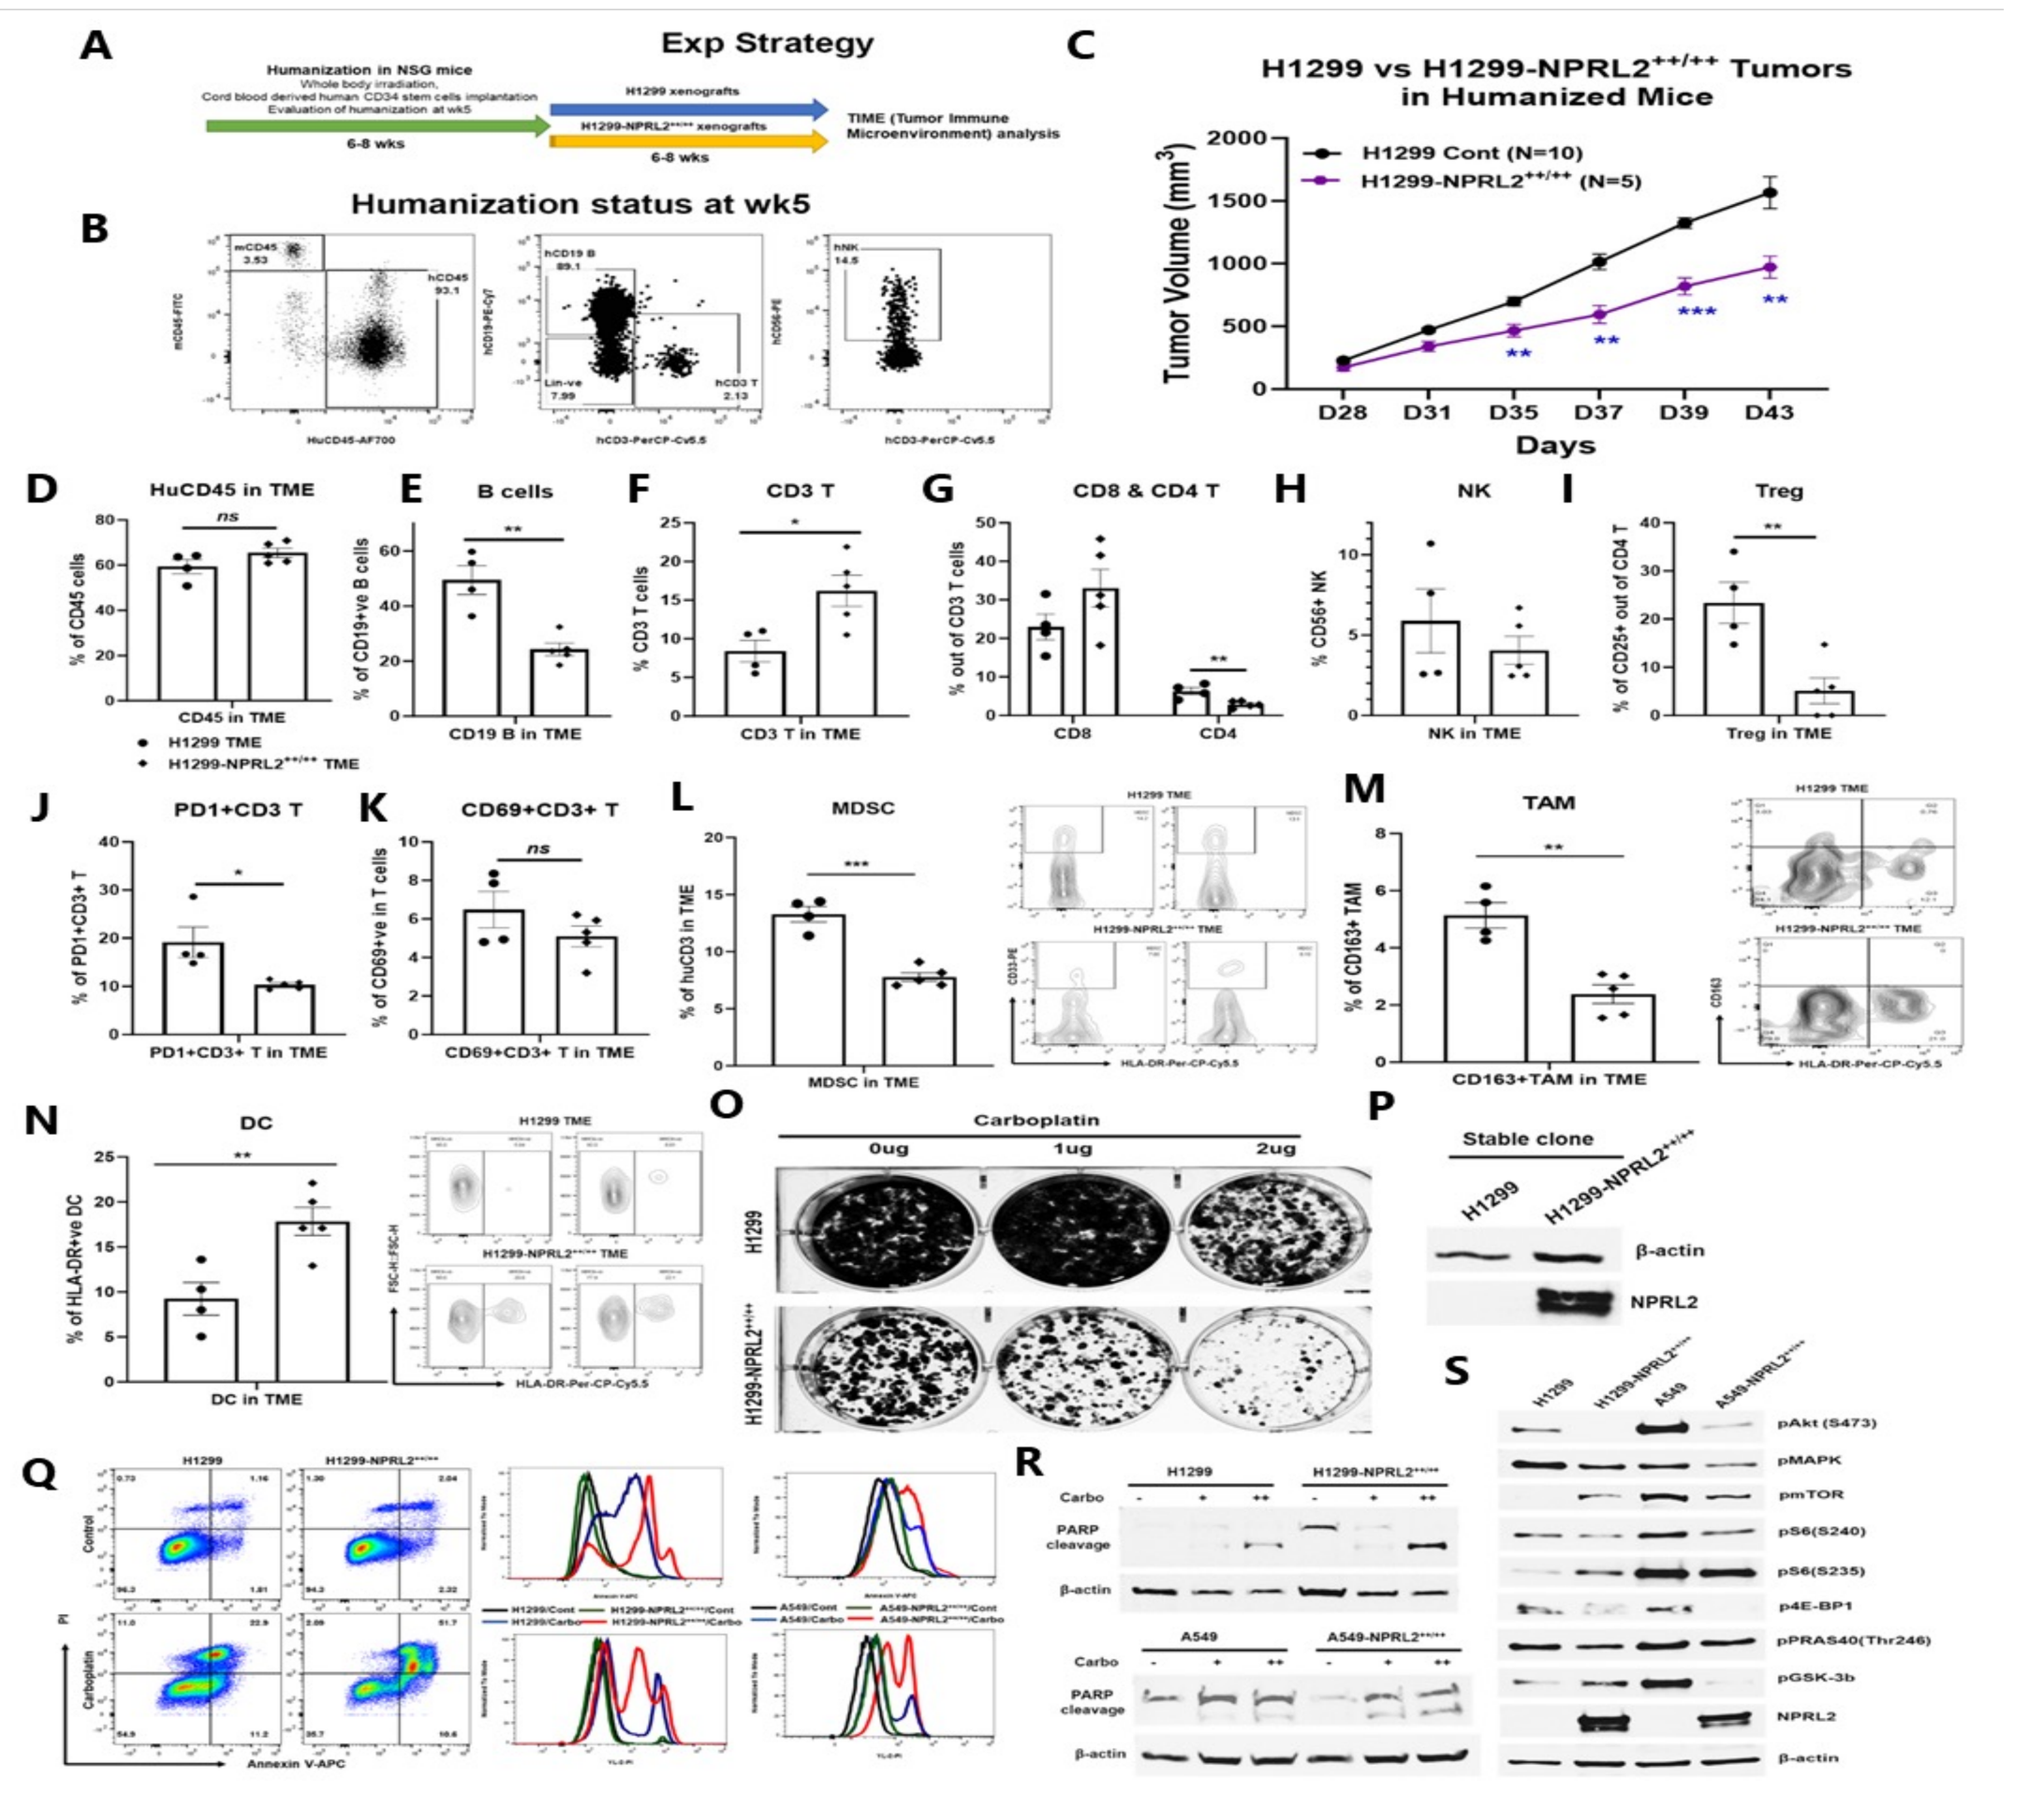

Fig 8. NPRL2 stable expression altered TME, induced apoptosis, inhibited cell growth and its signaling. A-C) Tumor growth comparison between NPRL2 stable vs control tumors in humanized mice; D-N) TME analysis of human CD45, B, CD3 T, CD4 T, CD8 T, NK, PD1+CD3 T, CD69+CD3 T, MDSC, TAM and DC in NPRL2 stable tumors; O) Inhibition of colony formation; P) NPRL2 expression in stable cells; Q) Carboplatin augmented apoptosis in NPRL2 stable cells; R-S) Altered level of PARP cleavage, pMAPK, and Akt-mTOR signaling molecules in NPRL2 stable clones.* means p < 0.05

Conclusions

- NPRL2 showed significant inhibition of colony formation and induced apoptosis in both KRAS^{wt}/aPD1^S H1299 and KRAS/STK11^{mt}/aPD1^R A549 NSCLC cells respectively.
- NPRL2 sensitivity was associated with upregulation of PD-L1 expression in the cells
- Liposomal delivery of NPRL2 gene therapy showed a significant antitumor effect on KRAS/STK11^{mt}/aPD1^R A549 lung metastasis in both humanized and non-humanized mice as well as KRAS^{mt}/aPD1^R LLC2 tumors in syngeneic mice.
- Greater antitumor effect was found in humanized mice vs non-humanized mice.
- No synergistic antitumor effect of NPRL2 + pembrolizumab combination was found on KRAS/STK11^{mt}/aPD1^R A549 and KRAS^{mt}/aPD^R LLC2 tumors, although a synergistic effect was found on KRAS^{wt}/aPD1^S H1299 tumors in humanized mice.
- The antitumor effect of NPRL2 was associated with increased infiltration of human CD45, CD3 T, cytotoxic T, NK cells, and fewer human regulatory T cells (Treg) in tumors.
- PD1 expressing exhausted CD8 T cells were downregulated whereas the number of activated T cells (CD69+CD8+T), effector (EM), and central memory (CM) CD8 T cells were significantly increased by NPRL2 treatment.
- NPRL2 significantly induced antigen presenting HLA-DR+ve dendritic cells and CD11c DC in tumors
- Depletion of CD8 T and Ag-presenting cells significantly attenuated the antitumor effect of NPRL2 on anti-PD1 resistant tumors.
- T cell functional genes, including IFNγ, CD8b, CD7, TNFSF18, ITGA1, GATA3, and TBX21 were significantly increased. Conversely, the negative regulatory genes, including FOXP3, TGFB1, TGFB2, and IL-10RA were inhibited in nanostring analysis.
- NPRL2 downregulated T cell co-inhibitory molecules, including CTLA4, ICOS, LAG3, PDCD1, CD274, IDO1, PDCD1LG2, CD47, and KLRB1.
- Stably expressing NPRL2 clones were established, and tumors in humanized mice with these clones exhibited altered TME which was associated with significantly slower growth compared to controls.
- NPRL2 stable cells showed a substantial increase in colony formation inhibition as well as heightened sensitivity to carboplatin in colony formation, apoptosis, and PARP cleavage assays.
- Stable expression of NPRL2 resulted in the downregulation of both AKT-mTOR and MAPK pathways by inhibition of pAKT, pmTOR, pPRAS40, p4E-BP1, pS6, and pERK1/2.

References & Disclosures

- Jayachandran G……Roth JA et al. NPRL2 sensitizes human non-small cell lung cancer (NSCLC) cells to cisplatin treatment by regulating key components in the DNA repair pathway, PLoS One, 2010 Aug 5;5(8). PMID: 20700484
- Ueda K,,,,,Roth JA et al. The 3p21.3 tumor suppressor NPRL2 plays an important role in cisplatin-induced resistance in human non-small-cell lung cancer cells. Cancer Res. 2006 Oct 1;66(19):9682-90. PMID: 17018626
- Jack A. Roth is a consultant, stock owner (including pending patent) in Genprex, Inc. All other authors have declared that no competing interests exist