CELsignia

Subgroup of ovarian cancer patients with hyperactive RAS network signaling identified: dynamic pathway activity test identifies patients that may benefit from PI3K/mTOR or PI3K/mTOR/BCL inhibitors

Stefano Rossetti, Aaron Broege, Joanna Sabat, Shu Wiley, Salmaan Khan, Catherine Kuzmicki, Ian MacNeil, Brian Sullivan, Lance Laing

Celcuity Inc., 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

celcuity

celcuity.com

Background

G-protein-coupled receptors (GPCRs) and their phospholipid ligands have well described links to cancer, including ovarian cancer (OC).[1,2] Lysophosphatidic acid (LPA) is a GPCR ligand with a long-known link to RAS/MAPK/PI3K oncogenic signaling. LPA can activate RAS that in turn activates PI3K-α to advance tumor growth.[3-5] Additionally, through several mechanisms, LPA can activate receptor tyrosine kinases (RTK) that in turn can work through PI3K-α or other PI3K isoforms to drive oncogenic signaling.[6-8] Due to the nonlinear, non-serial nature of the RAS signaling network and other complexities, identifying RAS nodes involved in oncogenic signal transduction has been challenging. Moreover, since inhibition of a single RAS node can trigger adaptation that results in activation of other RAS nodes, multiple RAS nodes and PI3K isoforms may need to be targeted to induce durable anti-tumor responses. To identify patients with dysregulated RAS signaling tumors who may respond to RAS node inhibitors, an assay using an impedance biosensor was developed. The CELsignia RAS Activity Test measures GPCR-initiated signaling activity and PI3K, mTOR, and BCL's role in transducing this activity in live tumor cells. In this test, LPA is used to stimulate multiple pathways linked to RAS activation and identify which of these RAS nodes are involved. The current study set out to characterize the prevalence of dysregulated RAS signaling initiated by LPA in OC patients and the role played by PI3K, mTOR and BCL.

Methods

Cell lines: The ovarian cancer cell lines used in this study were maintained according to ATCC recommendations and authenticated by ATCC.

Tissue specimens and patient tumor culture: A set of de-identified excess ovarian cancer tissue specimens was obtained from 32 patients. Methods for tumor cell extraction and culture were based on the Huang, et al. method.[9-10]

Sequencing: Genomic DNA was provided to contract research organizations for targeted next-generation sequencing to an average depth of 1000x. Sequencing was targeted to panels of 500–1000 genes frequently mutated in solid tumors with enrichment (Nimblegen SeqCap or Novogene NovoPM 1.0). Reads were aligned to the reference genome with BWA.

Flow cytometry: Flow cytometry on disaggregated tissue and cultured cells was performed on the Agilent Novocyte 3005. Antibody staining was performed by standard procedures. DNA staining for DNA index calculation was performed with FxCycle™ violet. Apoptosis and proliferation were assessed by staining with anti-Cleaved-Caspase 3 and by EdU incorporation assay, respectively. mTOR activity was assessed by staining with anti-RPS6 (pS235/S236).

CELsignia analysis: Dynamic live cell response to a GPCR agonist (LPA), a PI3K-α inhibitor (GDC-0077), a pan-PI3K/mTOR inhibitor (gedatolisib) and a BCL inhibitor (navitoclax) was measured using an xCELLigence impedance biosensor (Agilent Technologies). From these responses, the gross amount of GPCR-initiated signaling and corresponding participation of PI3K-α, all Class 1 PI3K-isoforms, mTORC1, and BCL, was quantified and converted to a signaling score.

Statistical analysis: A data set of 49 CELsignia LPA scores from OC cell lines and patient tumor cultures was analyzed. A normal mixture model was fitted to the combined data set using the normalmixEM procedure in the R package mixtools. Two runs of the statistical analysis were made, fitting 2 and 3 components, along with a baseline single-component model.

Figure 1: CELsignia analysis uses biosensor to quantify signaling activity in real time in live cells

- CELsignia analysis leverages connections among cell adhesion, impedance, and cell signal transduction
- Live cells are attached via ECM to a microelectrode on the bottom of a 96-well impedance biosensor plate. Additionally, the tumor epithelial cells in the wells form adhesion-based gap junctions
- The cells attached to the biosensor impede the flow of electrons when mVAC current is applied and changes in impedance (mΩ) are recorded
- Signaling activity causes cell adhesion changes that affect impedance levels recorded by the biosensor

Example Impedance vs. Time Data Set

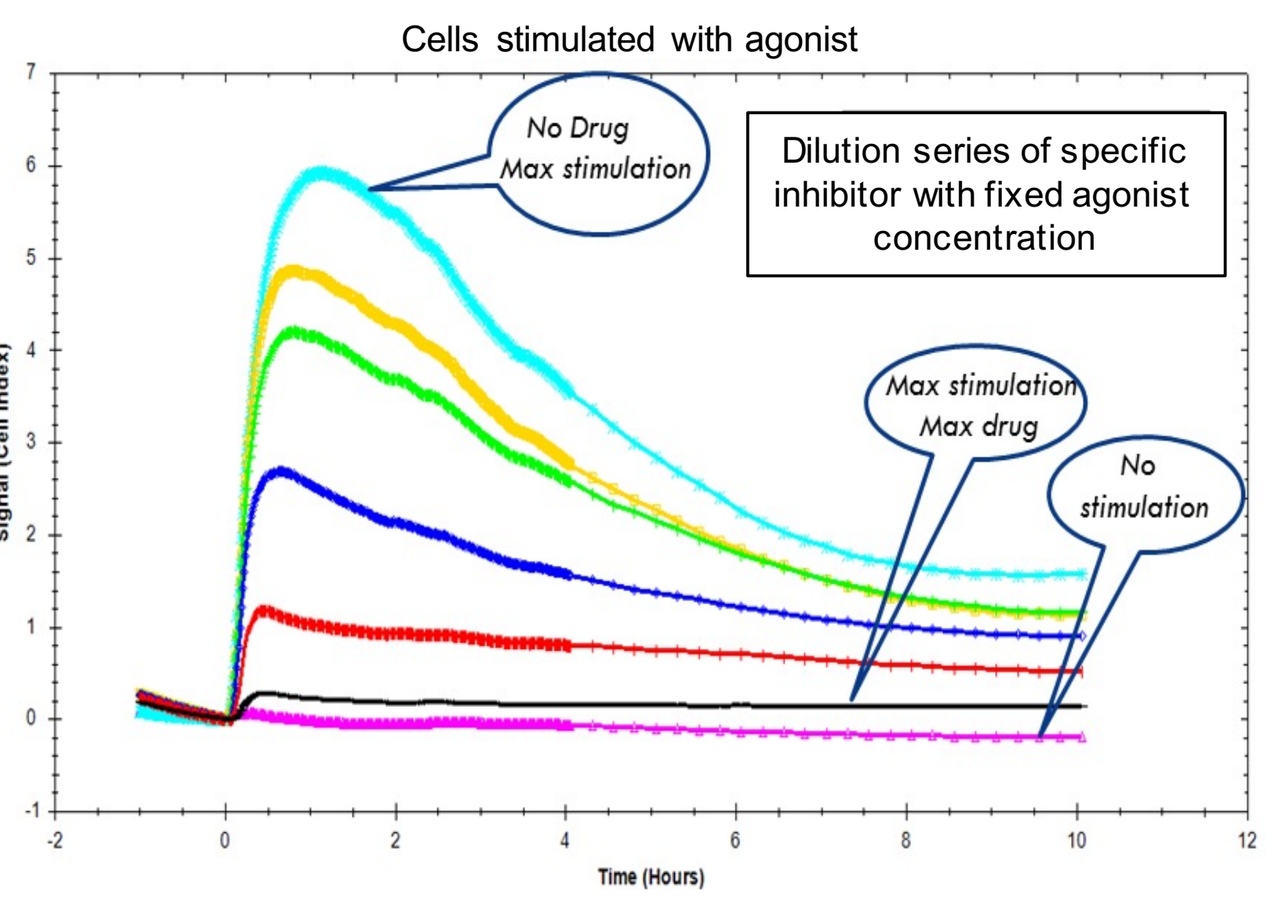

Cells stimulated with agonist
No Drug
Max stimulation
Dilution series of specific inhibitor with fixed agonist concentration
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Pathway specific ligands and inhibitors used to turn on/off signaling pathways

Very subtle cell adhesion changes are measured

Response quantified as signaling units over time to characterize signaling activity

Determine Signaling Score for each pathway tested

Results

Figure 2: LPA initiated activity measured by CELsignia in OC cell lines

A

Cell line	PIK3CA	PTEN	AKT1	KRAS	BRAF	TP53	CELsignia LPA response	LPAR1	LPAR2	LPAR3
	Mutational status							LPAR expression (CCLE)		
CaOV-3	wt	wt	wt	wt	wt	Mut	1218	4	10	7
UWB1.289	wt	wt	wt	wt	wt	Mut	782	NA	NA	NA
MES-OV	NA	NA	NA	NA	NA	NA	481	NA	NA	NA
OVCAR-3	wt	wt	wt	wt	AS	Mut	417	2	7	8
COV362	wt	wt	wt	wt	wt	Mut	251	9	2	0
TOV-21G	Mut	Mut	wt	Mut	wt	wt	244	11	3	0
SK-OV-3	Mut	wt	wt	wt	wt	Mut	181	5	7	2
HEY-T30#	wt	wt	wt	Mut	Mut	wt	181	NA	NA	NA
OVCAR-4	wt	wt	wt	wt	wt	Mut	86	0	6	1
OV-90	wt	wt	wt	wt	Mut	Mut	78	0	6	0
TOV-112D	wt	wt	wt	wt	wt	Mut	23	36	5	5
KURAMOCHI	wt	wt	wt	wt	wt	Mut	18	2	3	10
OVSAHO	wt	wt	wt	wt	wt	Mut	18	1	9	6
OVKATE	wt	wt	wt	wt	wt	Mut	1	1	6	0
CaOV-4	wt	wt	wt	wt	wt	Mut	-4	1	7	14
UACC2727	NA	NA	NA	NA	NA	NA	-6	NA	NA	NA
OVMANA	Mut	wt	wt	wt	wt	wt	-13	0	6	0
UACC-1598	NA	NA	NA	NA	NA	NA	-23	NA	NA	NA

B

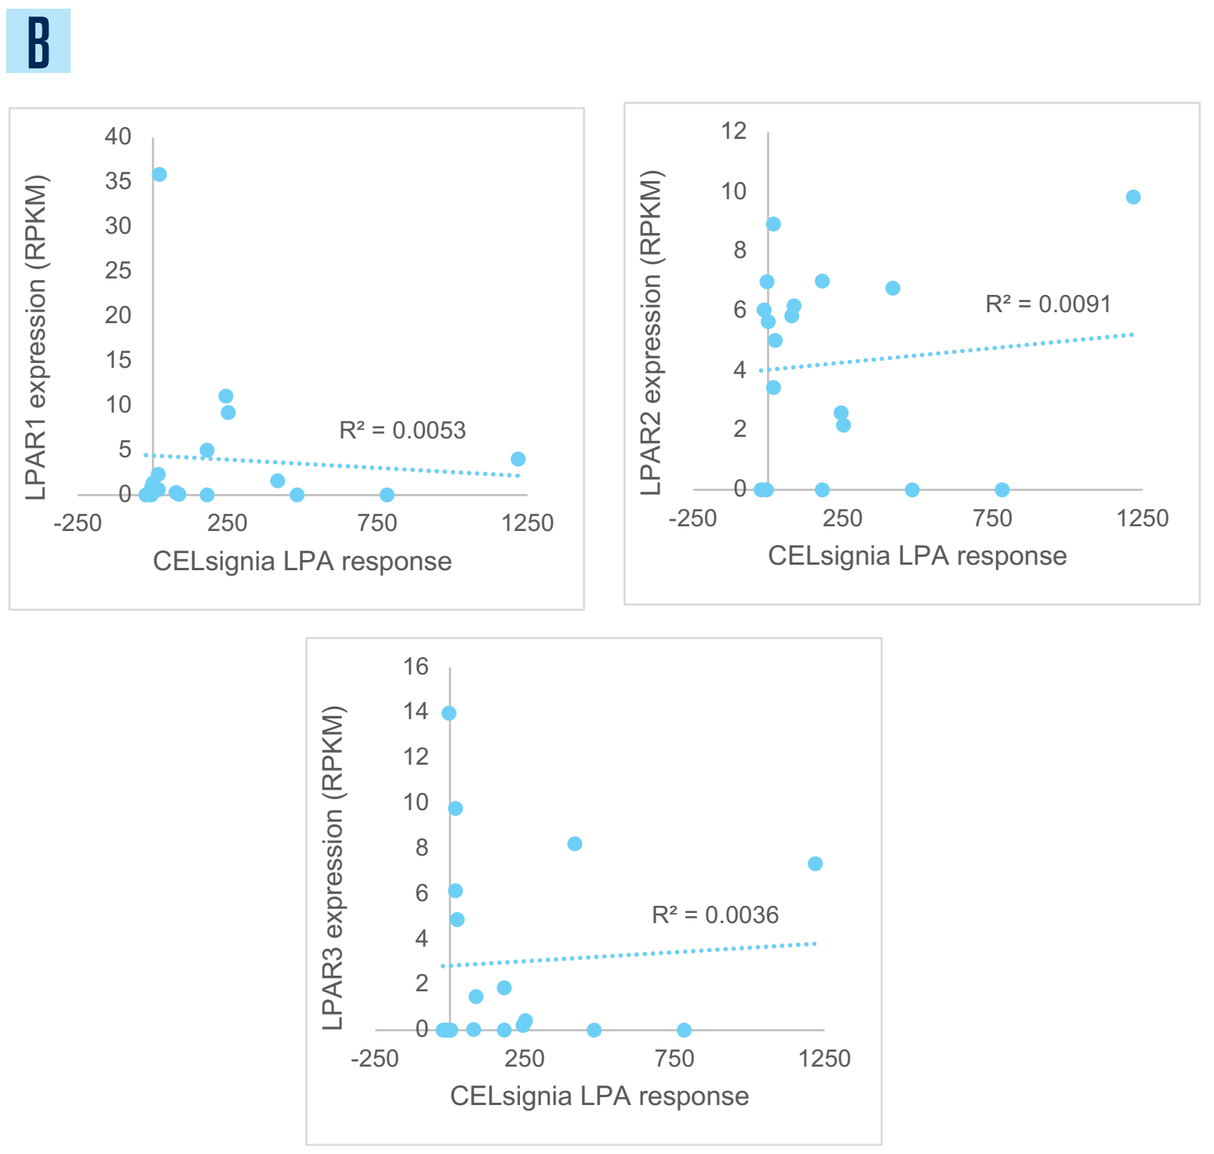

LPAR1 expression (RPKM)
R^2 = 0.0053
CELsignia LPA response
LPAR2 expression (RPKM)
R^2 = 0.0091
CELsignia LPA response
LPAR3 expression (RPKM)
R^2 = 0.0036
CELsignia LPA response

(A) Table showing the mutational status, CELsignia LPA response and LPA receptors (LPAR1-3) expression in a panel of 18 OC cell lines. LPAR expression data show RPKM from the Cancer Cell Line Encyclopedia (CCLE). Conditional formatting was applied to all LPARs columns. NA = not available; mut = mutated; wt = wild type. **(B)** Scatter plots showing that lack of correlation between CELsignia LPA response and LPAR1, LPAR2, and LPAR3 expression.

These results show that:

1. **LPA signaling response in OC cells does not correlate with overexpression of LPA receptors or mutations in key RAS signaling pathway genes**

Figure 3: CELsignia analysis of RAS signaling in OC patient cultures

A C1113—3-day culture

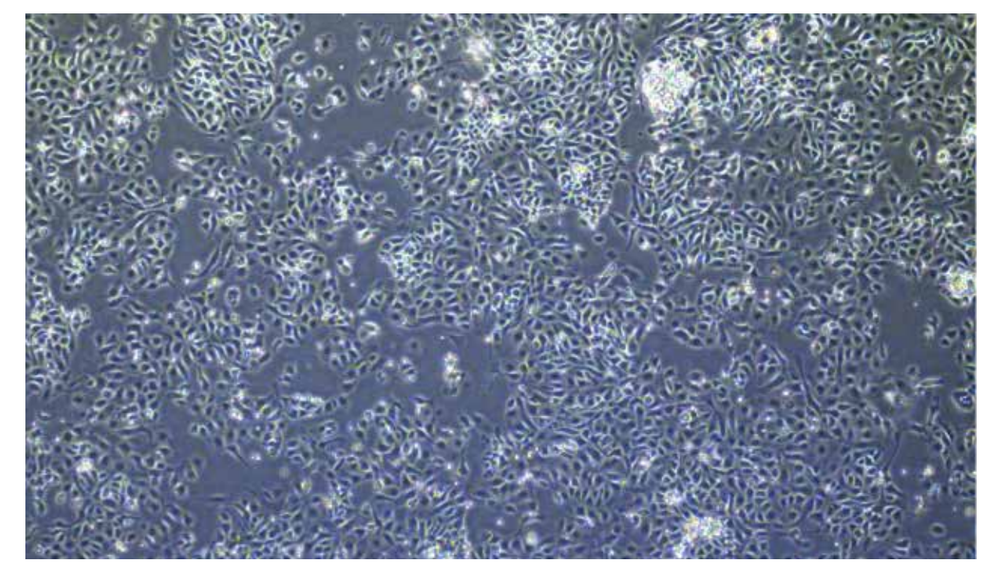

B

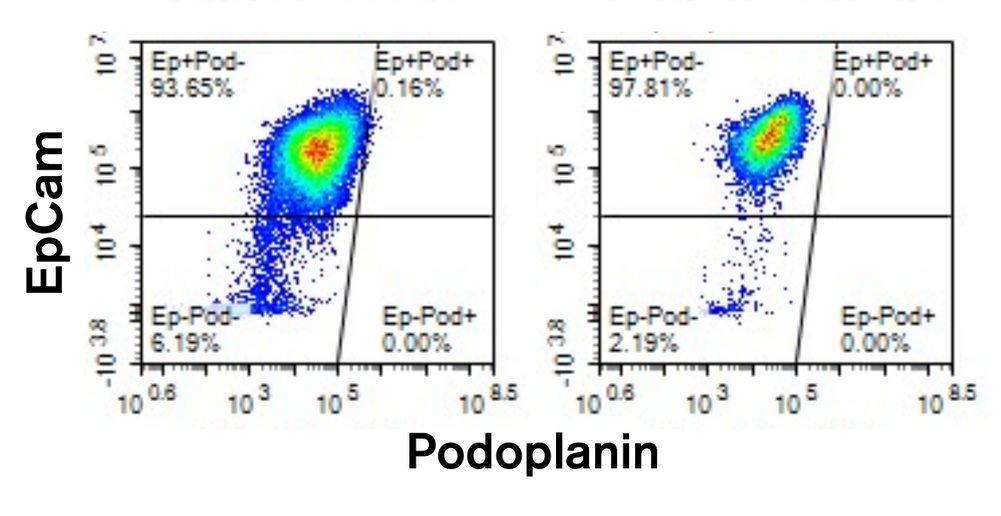

C1113
Tumor tissue
C1113
Patient culture
EpCam
Podoplanin
Ep+Pod-
93.65%
Ep+Pod+
0.16%
Ep-Pod-
6.19%
Ep-Pod+
0.00%
Ep+Pod-
97.81%
Ep+Pod+
0.00%
Ep-Pod-
2.19%
Ep-Pod+
0.00%

C

Gene	Mutation	Prediction	Variant Allele Frequency: Normal tissue	Tumor tissue	Tumor culture
TP53	P87fs*	pathogenic	0	0.9	0.86
MSI2	V201M	pathogenic	0	0.89	0.88
TET2	Y1315H	-	0	0.93	0.89
ARID1A	D1354N	pathogenic	0	0.43	0.44
IGF2R	G601A	-	0.01	0.91	0.85

D

Sample	Subtype	Age	Ethnicity	Stage	FACS analysis % EpCam+ cells: Tissue	Culture	FACS analysis DNA index in EpCam+ cells: Tissue	Culture	CELsignia LPA response: Culture
C1944	HGS	52	Caucasian	pT3c N1	65%	95%	1.28	1.59	792
C1966	HGS	58	Caucasian	pT3c N1	97%	94%	1.03	1.03	746
C1735	HGS	62	Caucasian	pT3a Nx	75%	90%	1.69	1.75	727
C2032	HGS	78	Caucasian	NA	75%	96%	1.42	1.68	679
C2131	Clear cell	60	Caucasian	pT2a N0	56%	99%	1.04, 1.79	1.09, 1.71	624
C1508	Endo-Met	48	Caucasian	pT3a Nx	50%	80%	1.01	1.09	502
C1928	HGS	67	African Am.	pT3b N0	80%	90%	1.56	2.00	264
C2332	HGS-Met	66	Caucasian	NA	51%	74%	1.54	1.64	238
C2096	HGS	63	Caucasian	pT3c Nx	78%	77%	1.73	1.86	204
C1877	Endom.	53	Caucasian	pT1b N0	43%	92%	1.02	1.31	182
C2117	Endom.	50	Caucasian	pT1c N0	58%	86%	1.30	1.40	178
C2408	HGS	59	Caucasian	pT1c3 N1a	30%	74%	1.72	1.84	161
C1504	HGS	58	African Am.	pT2 N0	71%	92%	1.65	1.70	161
C2155	HGS	78	Caucasian	NA	81%	96%	1.37	1.69	146
C2040	HGS	49	Asian	pT3c N1	98%	83%	0.86	0.98	118
C1113	HGS	51	Caucasian	pT3c N1	97%	98%	1.57	1.65	91
C2061	HGS-Met	70	Caucasian	pT3c N1	31%	88%	1.27	1.36	70
C2358	HGS-Met	81	Caucasian	pT3c Nx	34%	78%	1.79	1.91	67
R2063	HGS-Met	74	Caucasian	NA	29%	78%	1.64	1.67	55
C1620	HGS-Met	42	Caucasian	NA	79%	87%	1.04	1.15	55
C1497	HGS	58	Caucasian	pT3c N1	80%	86%	1.29	1.38	41
C1468	HGS	65	Caucasian	pT3c N1	29%	78%	1.24	1.40	19
C2427	HGS-Met	48	Caucasian	pT3c NX	78%	92%	1.37	1.51	14
C2022	HGS-Met	67	Caucasian	pT3b Nx	42%	71%	0.92	1.16	12
C2457	HGS-Met	64	Caucasian	pT3c Nx	82%	76%	1.31	1.76	12
C2064	HGS-Met	70	Caucasian	NA	85%	98%	1.48	1.75	10
C2174	HGS	60	Caucasian	pT3c Nx	31%	71%	1.31	1.42	10
C1774	HGS	48	Caucasian	pT3c N1	74%	89%	1.35	1.66	7
C1693	Endom.	69	Caucasian	pT1c N0	91%	97%	1.40	1.37	2
C2197	HGS-Met	58	Caucasian	NA	89%	80%	0.98	1.09	2
C2060	HGS	59	Caucasian	pT3c N0	77%	91%	0.93	1.21	-2

(A) Representative culture from a digested OC patient tumor biopsy showing epithelial cells with a tight cobblestone structure. **(B)** Representative FACS analysis of an initial disaggregated tissue and the resulting cultured cells showing similar % of epithelial (EpCam+) and stromal cells (EpCam-/Podoplanin+). **(C)** Genetic analysis confirms that cultured cells are enriched for mutations present in cancer tissue (selected mutations are shown). **(D)** Table showing the CELsignia LPA response in patient cultures derived from 31 OC patients' tumor tissues. The cultures were enriched for epithelial (EpCam+) cells with a DNA index similar to the one found in the original tumor tissue. See Figure 4 for cutoff of CELsignia LPA response.

These results show that:

1. **Patient tumor cultures are enriched for epithelial cancer cells present in the original OC tissue**
2. **7/31 OC patient cultures have a CELsignia LPA response >250, which indicates hyperactive RAS signaling activity**

Figure 4: Estimating LPA signal cutoff

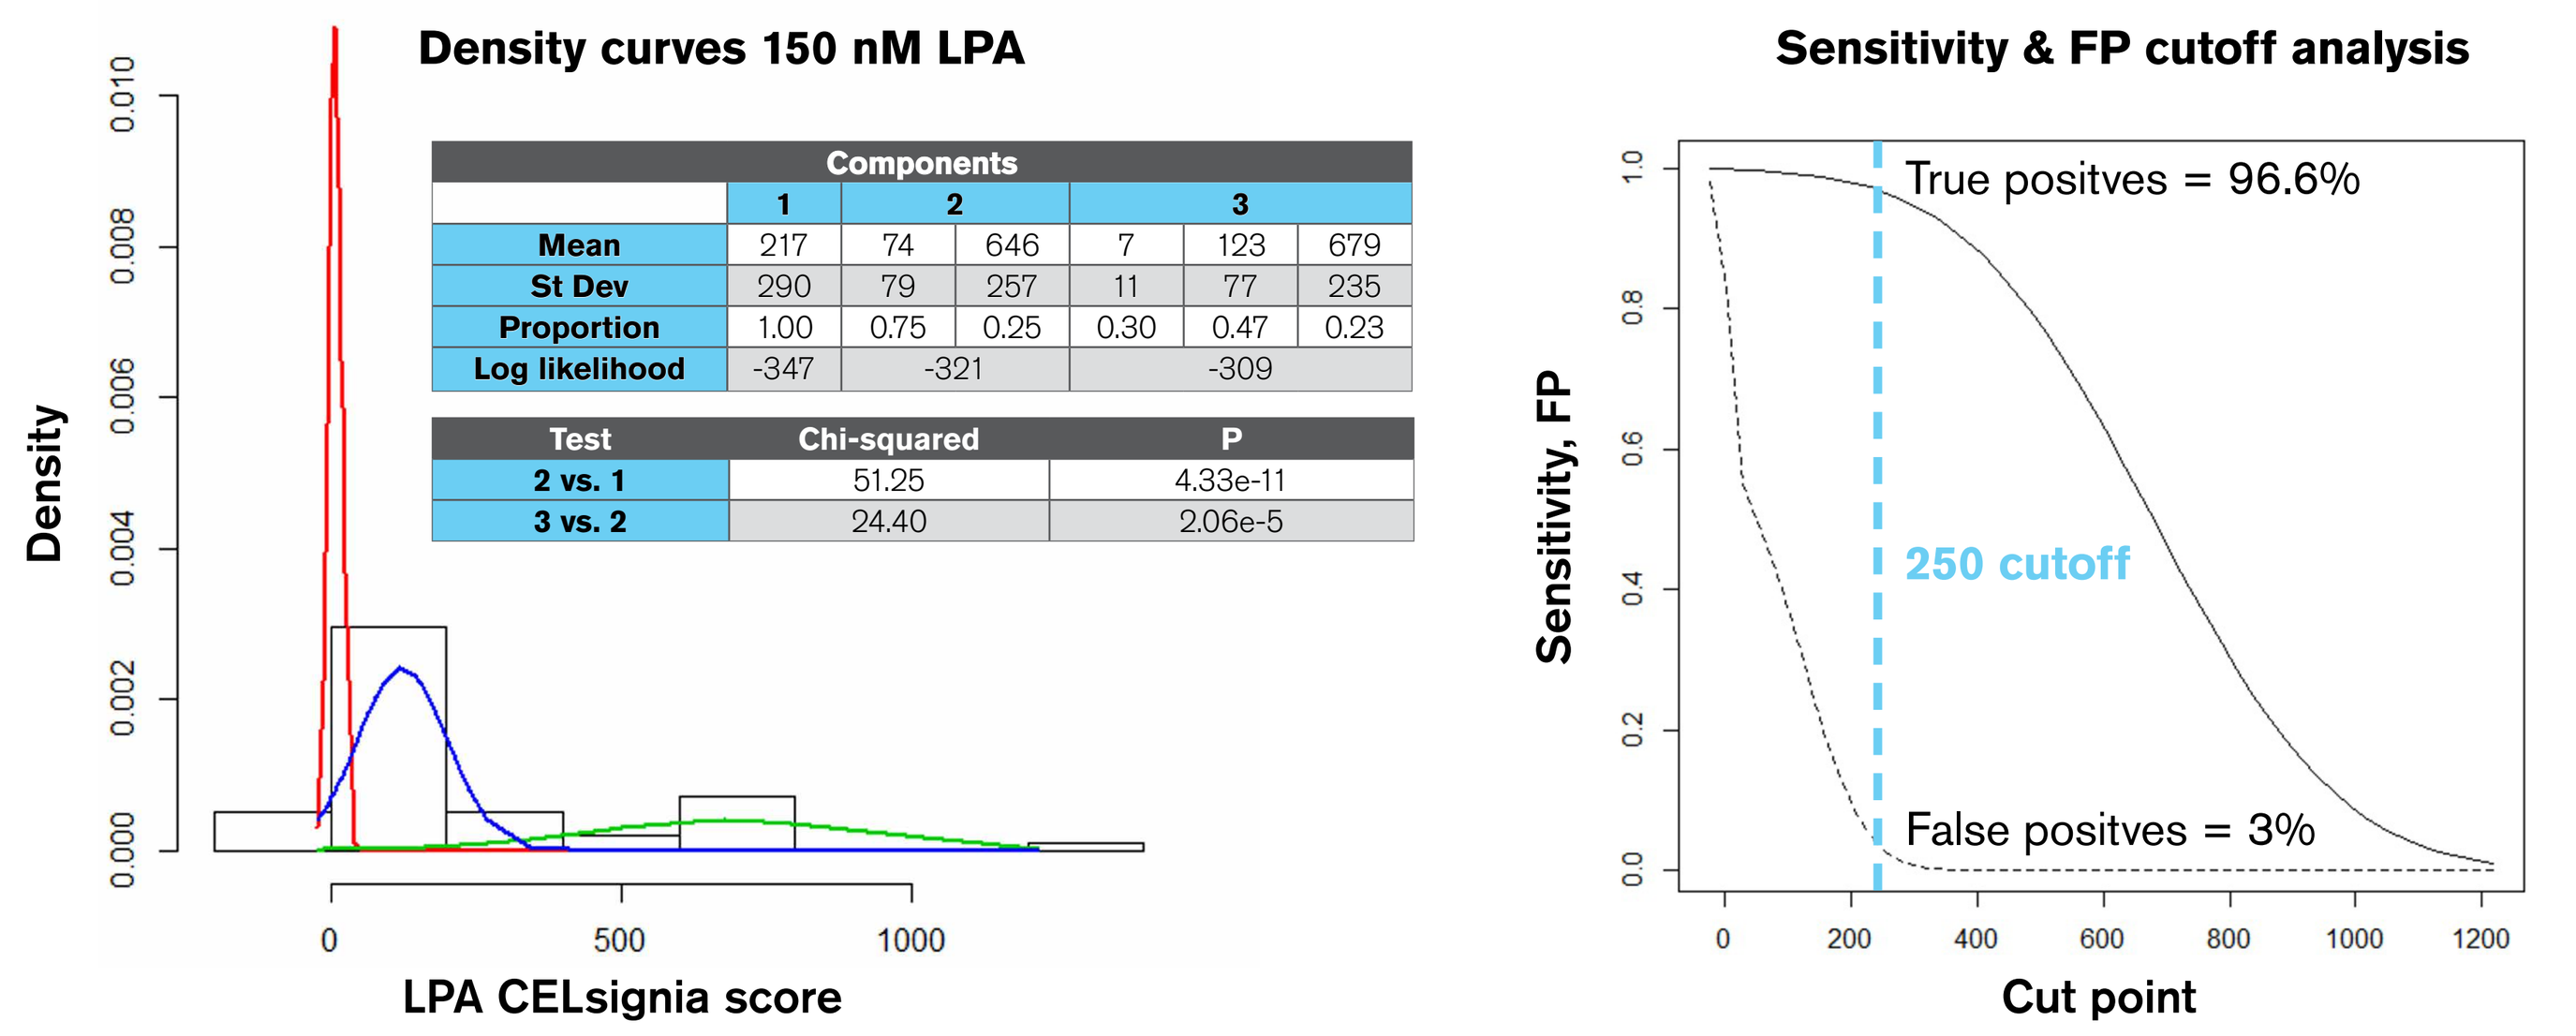

Density curves 150 nM LPA
Density
LPA CELsignia score
Sensitivity & FP cutoff analysis
True positves = 96.6%
250 cutoff
False positves = 3%
Sensitivity, FP
Cut point

	Components: 1	2		3		
Mean	217	74	646	7	123	679
St Dev	290	79	257	11	77	235
Proportion	1.00	0.75	0.25	0.30	0.47	0.23
Log likelihood	-347	-321		-309		

Test	Chi-squared	P
2 vs. 1	51.25	4.33e-11
3 vs. 2	24.40	2.06e-5

The charts and relative tables show the statistical analysis of the LPA CELsignia scores from 18 OC cells lines and 31 OC patient cultures. The good components separation in response to LPA is a prerequisite for determining test cutoffs in response to antagonists of specific RAS-related signaling pathways (e.g., the PI3K/mTOR inhibitor gedatolisib).

These results show that:

1. **A test score cutoff of 250 for LPA signaling can separate OC patients into two distinct populations (component 3 vs. components 1 & 2), where component 3 has abnormally high RAS signaling**
2. **A test score cutoff of 250 has >96% sensitivity and 3% false positive rate**
3. **Using the 250 cutoff, <u>12/49 (24%)</u> of OC cell lines and OC patient cultures have hyperactive RAS signaling**

Figure 5: Hyperactive RAS signaling involves the PI3K, mTOR, & BCL nodes

A

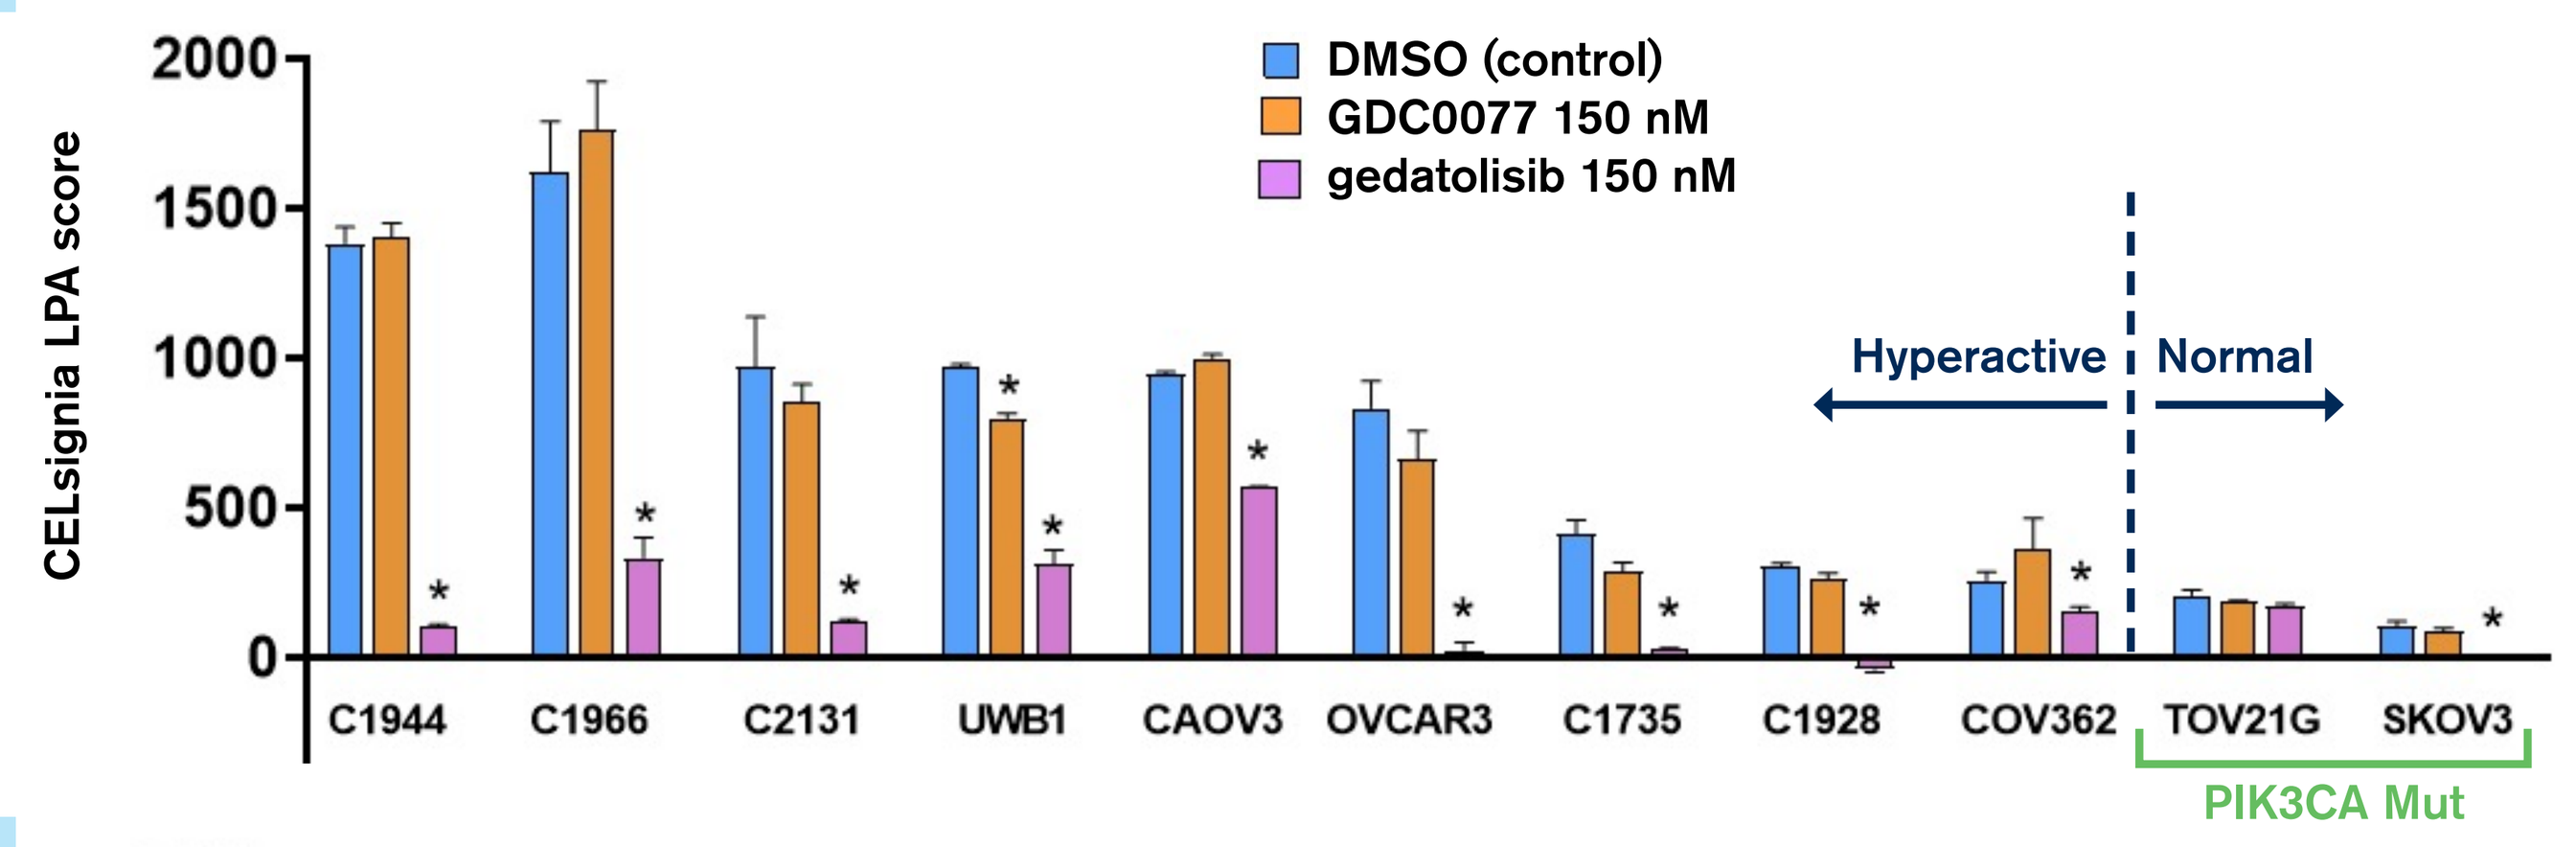

DMSO (control)
GDC0077 150 nM
gedatolisib 150 nM
CELsignia LPA score
Hyperactive
Normal
C1944
C1966
C2131
UWB1
CAOV3
OVCAR3
C1735
C1928
COV362
TOV21G
SKOV3
PIK3CA Mut

B

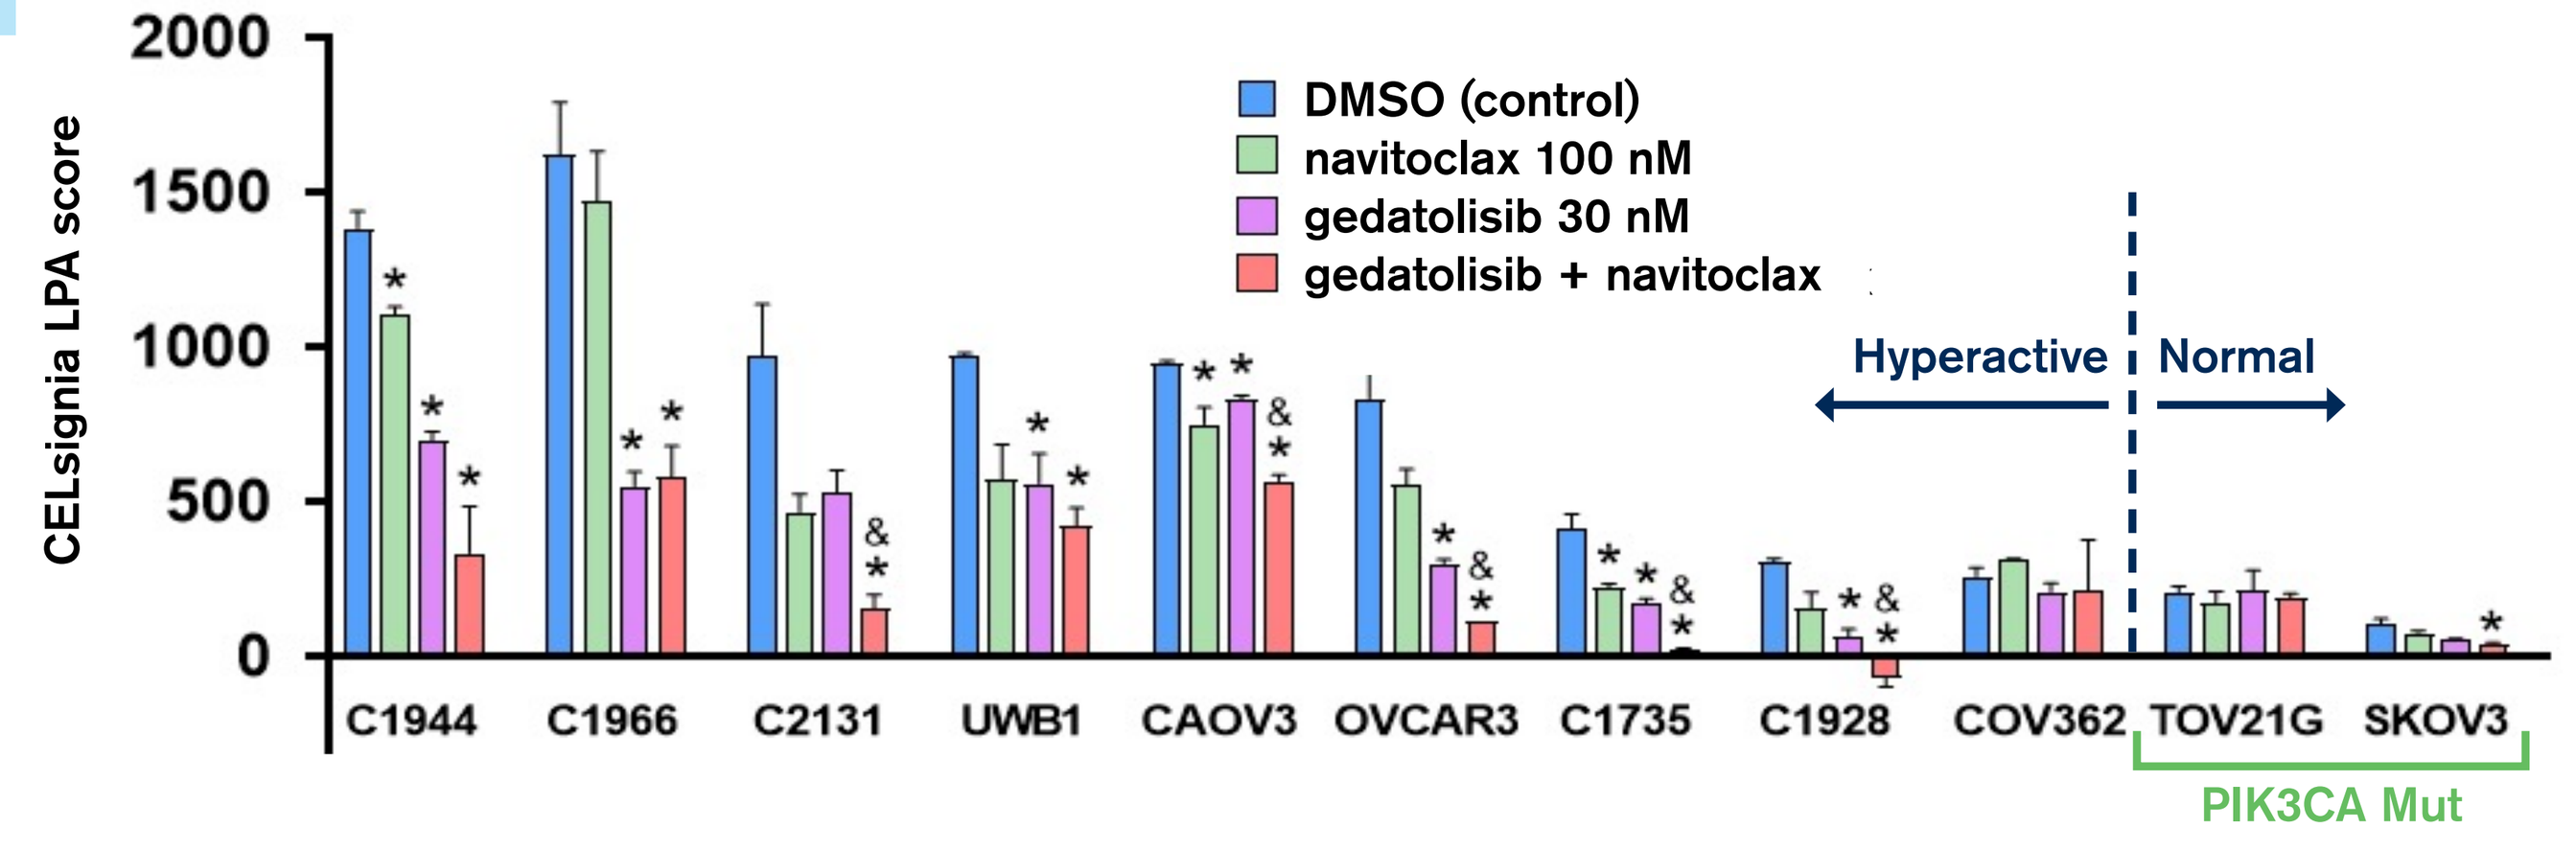

DMSO (control)
navitoclax 100 nM
gedatolisib 30 nM
gedatolisib + navitoclax
CELsignia LPA score
Hyperactive
Normal
C1944
C1966
C2131
UWB1
CAOV3
OVCAR3
C1735
C1928
COV362
TOV21G
SKOV3
PIK3CA Mut

Charts showing the response of 9 OC cell lines and patient cultures with hyperactive RAS signaling and 2 OC cell lines with normal RAS signaling to a PI3K-α inhibitor (GDC-0077), a pan-PI3K/mTOR inhibitor (gedatolisib) **(A)** and gedatolisib with a BCL inhibitor (navitoclax) **(B)**. Two concentrations of gedatolisib were selected to isolate and measure the pan-PI3K/mTOR signal, as well as measure the effect of combining with BCL inhibitor, navitoclax* indicates $p<0.05$ relative to DMSO control; & indicates $p<0.05$ relative to gedatolisib. TOV21G and SKOV3 carry the H1047Y and H1047R PI3K-α mutation, respectively.

These results show that:

1. **RAS hypersignaling initiated by LPA primarily involves the PI3K and mTOR nodes**
2. **Attenuation of LPA hypersignaling with a PI3K-α inhibitor was less effective than a PI3K/mTOR inhibitor**
3. **RAS hypersignaling may involve cooperation between BCL and the PI3K and mTOR nodes in 5/9 (56%) patients**
4. **The presence of PI3K-α mutations does not predict the response to GDG0077, gedatolisib or gedatolisib+ navitoclax**

Figure 6: The effects of PI3K, mTOR, and BCL inhibitors on CELsignia RASs+ tumors correlate with changes in cell physiology markers

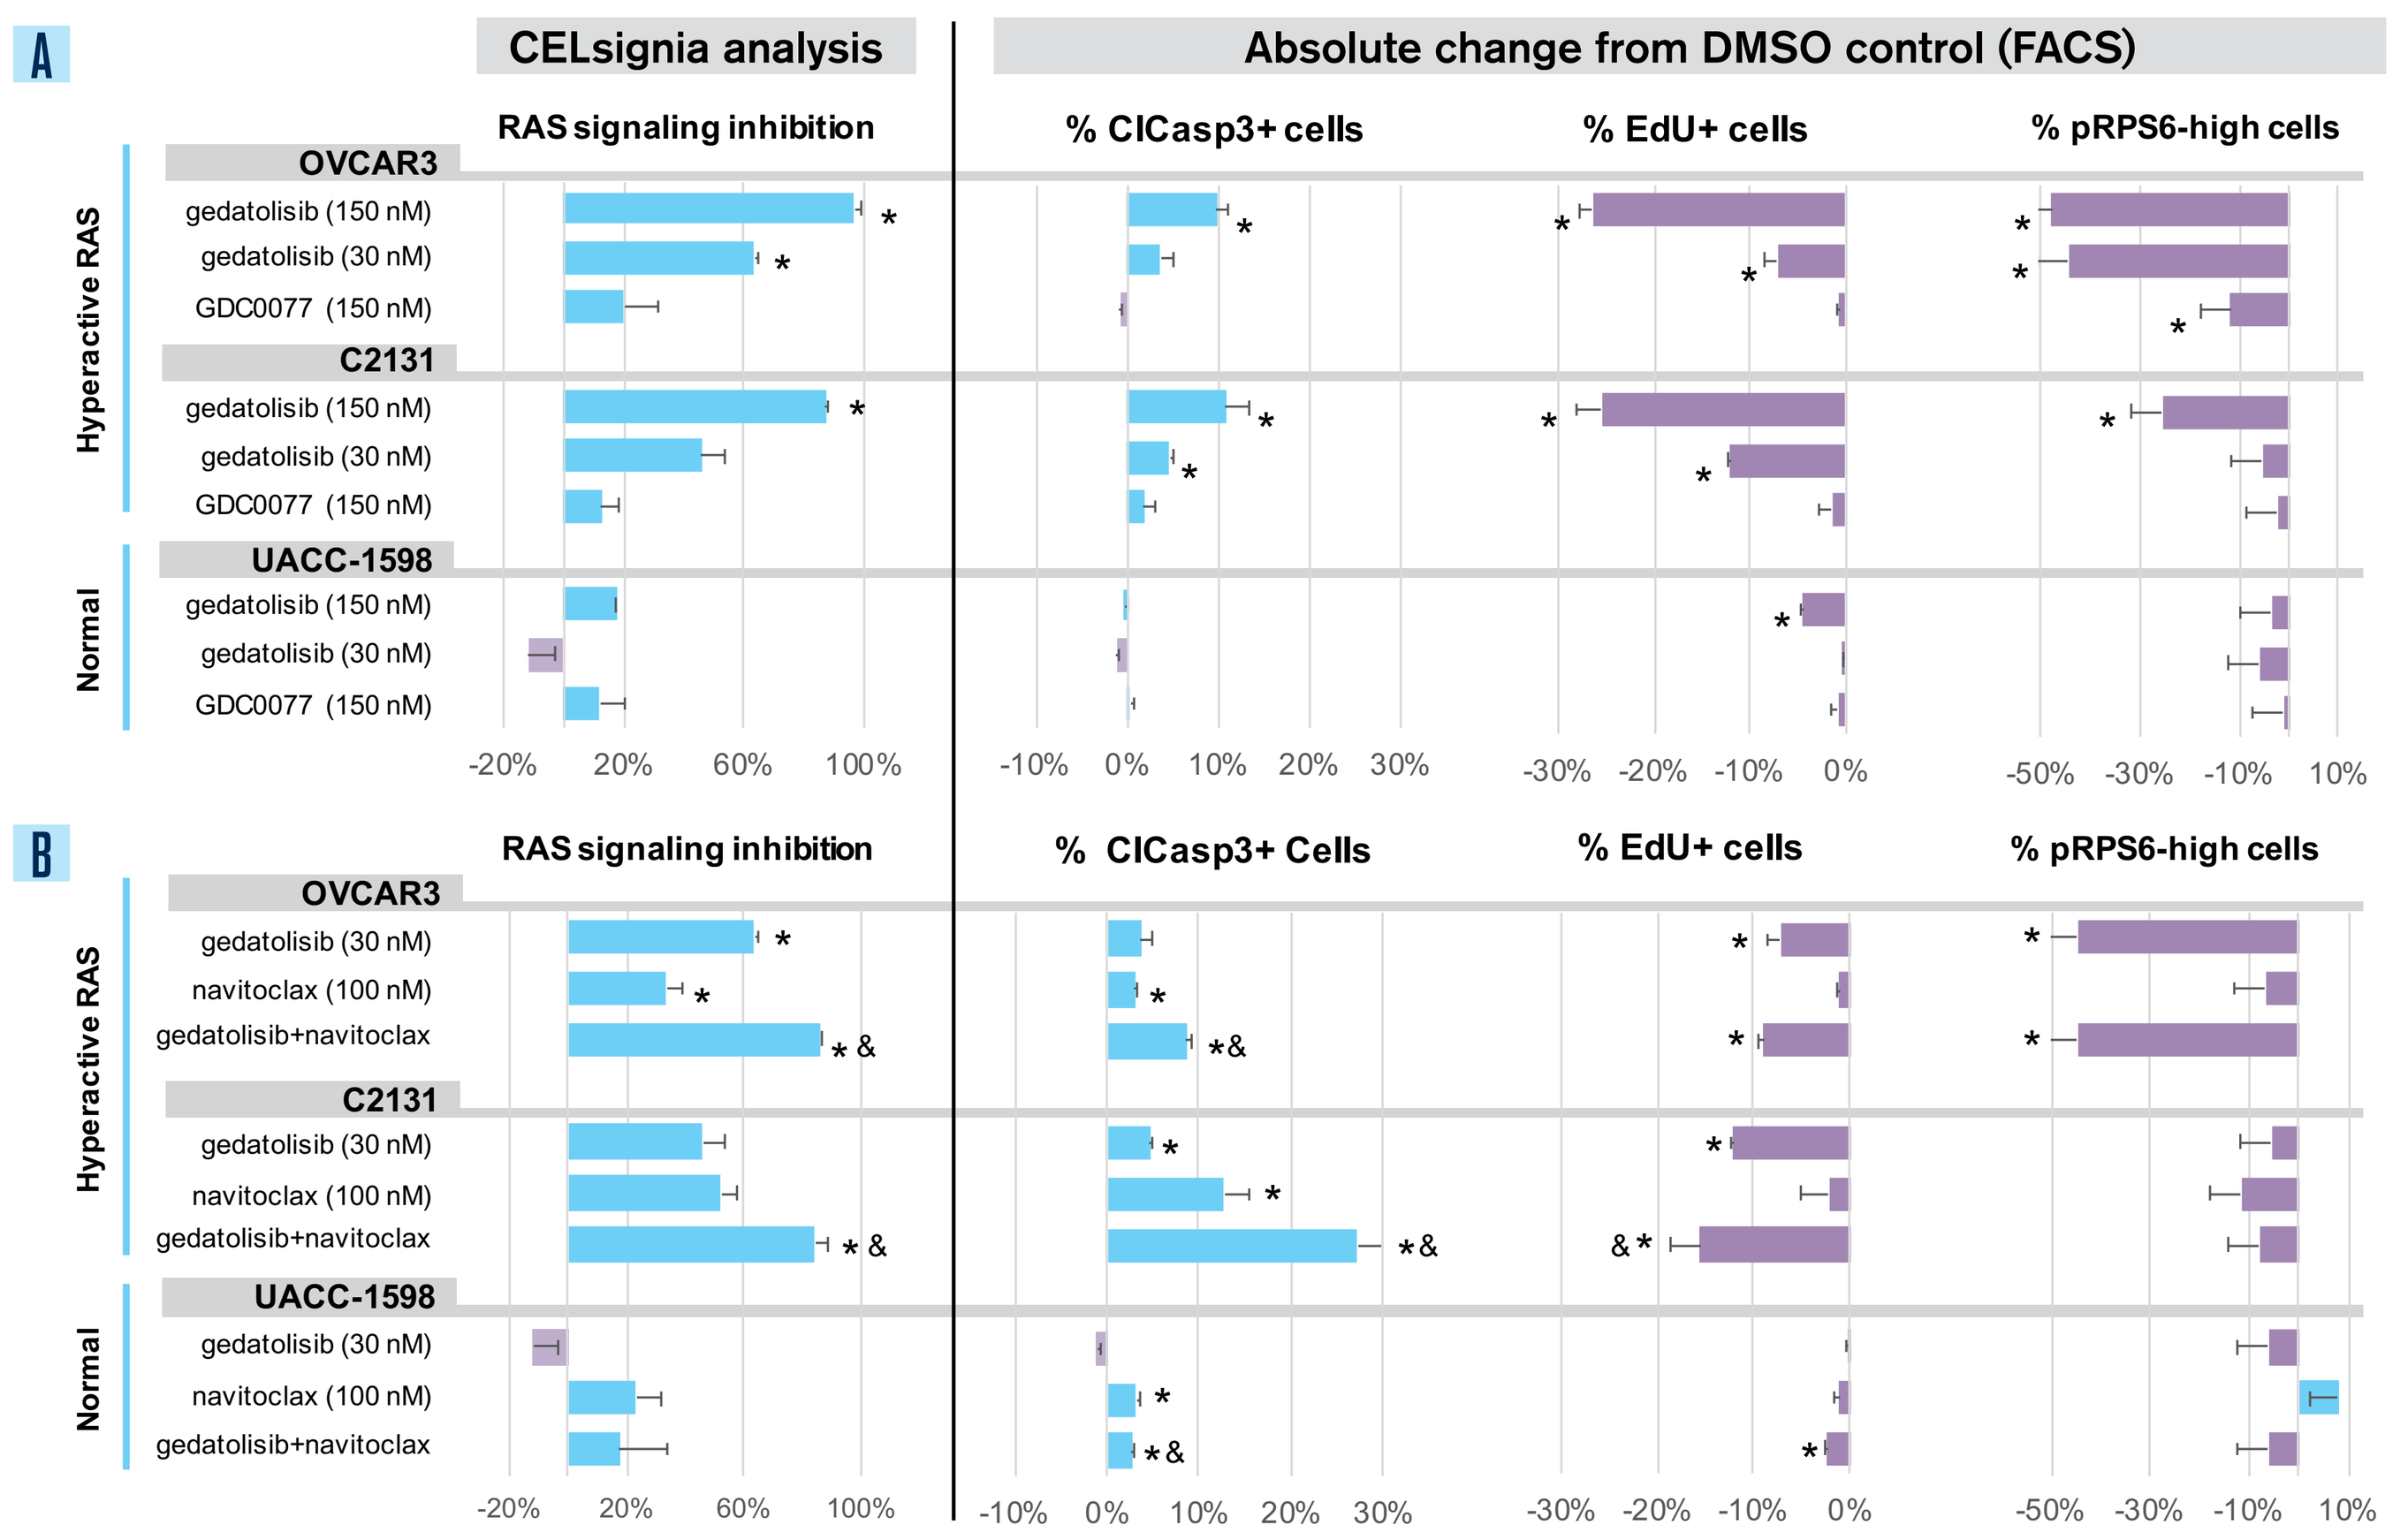

A
CELsignia analysis
Absolute change from DMSO control (FACS)
RAS signaling inhibition
% ClCasp3+ cells
% EdU+ cells
% pRPS6-high cells
Hyperactive RAS
Normal
OVCAR3
gedatolisib (150 nM)
gedatolisib (30 nM)
GDC0077 (150 nM)
C2131
UACC-1598
B
% ClCasp3+ Cells
gedatolisib (30 nM)
navitoclax (100 nM)
gedatolisib+navitoclax

CELsignia LPA response and FACS analysis of apoptosis (assessed by cleaved caspase 3), proliferation (assessed by EdU incorporation), and mTOR activation (assessed by pRPS6) in OC cell lines and patient cells treated with **(A)** either gedatolisib or GDC0077 or **(B)** gedatolisib, navitoclax, and gedatolisib + navitoclax. The results with these markers correlate with the CELsignia LPA response in UACC-1598, which had normal RAS signaling, relative to OVCAR3 and C2131, which had hyperactive RAS signaling, when assessed with gedatolisib (150 nM) or the combination of gedatolisib (30nM) + navitoclax (100 nM).

These results provide evidence that hyperactive RAS signaling detected by the CELsignia RAS test is oncogenic.

1. **Consistent with the CELsignia analysis, inhibiting PI3K/mTOR with gedatolisib induces more apoptosis and reduces proliferation more than inhibiting PI3K-α with GDC-0077**
2. **The greater level of apoptosis induced when BCL and PI3K/mTOR inhibitors are combined is consistent with the synergy found between BCL and PI3K/mTOR signaling by the CELsignia analysis**

Summary of Results

- The CELsignia RAS Activity Test identified patients with hyperactive RAS signaling regardless of LPAR expression of mutational status of RAS signaling-related genes
- The CELsignia RAS Activity Test identified 12 of 49 (24%) OC patients with hyperactive RAS signaling
- Hyperactive RAS signaling is always more effectively inhibited with a PI3K/mTOR inhibitor (gedatolisib) than a PI3K-α inhibitor (GDC-0077)
- More complete attenuation of hyperactive RAS signaling occurred in more than 50% of patients when PI3K, mTOR and BCL were simultaneously inhibited
- The attenuation of hyperactive RAS signaling by gedatolisib or gedatolisib+navitoclax correlates with reduced proliferation, reduced mTOR signaling and induction of apoptosis

Conclusions

These findings suggest that a significant subgroup of OC patients have a RAS-involved oncogenic driver that is responsive *ex vivo* to pan-PI3K/mTOR and pan-PI3K/mTOR + BCL inhibitors. A clinical trial to evaluate treatment response of this patient subgroup to combined PI3K/mTOR or PI3K/mTOR + BCL inhibitors is warranted.

References

1. Bar-Shavit, et al. G protein-coupled receptors in cancer. *Int J Mol Sci.* 2016;17(8):1320.
2. Predescu, et al. G protein-coupled receptors (GPCRs)-mediated calcium signaling in ovarian cancer. *Int J Mol Sci.* 2019;20(22):5568.
3. Rubio, et al. RAS activation in response to lysophosphatidic acid requires a permissive input from the epidermal growth factor receptor. *Biochem J.* 2003;376(Pt 3):571-576.
4. Downward. Role of receptor tyrosine kinases in G-protein-coupled receptor regulation of RAS: transactivation or parallel pathways? *Biochem J.* 2003;376(Pt 3):e9-10.
5. Rodriguez-Viciana, et al. Activation of phosphoinositide 3-kinase by interaction with RAS and by point mutation. *EMBO.* 1996;15(10):2442-2451.
6. Herrlich, et al. Ligand-independent activation of platelet-derived growth factor receptor is a necessary intermediate in lysophosphatidic, acid-stimulated mitogenic activity in L cells. *PNAS.* 1998;95(15): 8985-8990.
7. Hu, et al. Lysophosphatidic acid induction of vascular endothelial growth factor expression in human ovarian cancer cells. *J Natl Cancer Inst.* 2001;93(10):762-768.
8. Fang, et al. Mechanisms for lysophosphatidic acid-induced cytokine production in ovarian cancer cells. *J Biol Chem.* 2004;279(10):9653-9661.
9. Huang, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016;7(48):78577-78590.
10. Huang, et al. Development of a test that measures real-time HER2 signaling function in live breast cancer cell lines and primary cells. *BMC Cancer.* 2017;17:199.

Abstract #: 367

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.